IDWEEK 2024 | Poster #P1363

Cefiderocol Activity against Clinical Enterobacterales Isolates Carrying Metallo-β-Lactamase Genes in United States and European Hospitals (2020–2023)

RE Mendes, JH Kimbrough, D Beekman, JM Maher, HS Sader, M Castanheira

Element Iowa City (JMI Laboratories), North Liberty, IA, USA

Introduction

- β-lactamase genes, such as those encoding class A and D serine carbapenemases and class B metallo-β-lactamases (MBL), contribute to the emergence and dissemination of carbapenem-nonsusceptible Enterobacterales.
- MBL-producing Enterobacterales have become endemic in numerous countries around the globe, especially bla_{NDM}-carrying organisms.
 - More recently, several reports described the emergence and dissemination of Enterobacterales carrying multiple carbapenemase genes.
- Cefiderocol is approved by the US Food and Drug Administration (FDA) for the treatment of complicated urinary tract infections, including pyelonephritis, as well as hospital-acquired bacterial pneumonia and ventilator-associated bacterial pneumonia.
 - Cefiderocol is also approved in Europe for the treatment of infections in adult patients caused by aerobic Gram-negative organisms with limited treatment options.
- Cefiderocol is a siderophore cephalosporin with broad activity against Gram-negative bacteria, including multidrug-resistant (MDR) organisms like carbapenem-resistant Enterobacterales (CRE), carbapenem-resistant *Pseudomonas aeruginosa*, and *Acinetobacter baumannii*.
- The antibacterial activity of this molecule is due to its ability to achieve high periplasmic concentrations by hijacking the bacterial iron transport machinery, which increases cell entry.
 - In addition, cefiderocol remains stable to hydrolysis by serine β-lactamases (ESBLs, KPCs, and OXA-type carbapenemases) and MBLs.
- In this study, cefiderocol and comparator activities were analyzed against Enterobacterales carrying MBL genes, as part of the SENTRY Antimicrobial Surveillance Program for the US and Europe during 2020–2023.

Materials and Methods

Bacterial organisms

- This study comprised a collection of 32,053 Enterobacterales collected from various clinical specimens from patients hospitalized in 35 medical centers in the US and 42 sites in Europe, including Turkey and Israel, during 2020–2023. Only consecutive isolates (1 per patient infection episode) responsible for documented infections according to local criteria were included.
- Bacterial identification was confirmed by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following the Clinical and Laboratory Standards Institute (CLSI) M07 (2018) guidelines.
- Frozen-form broth microdilution panels were manufactured by JMI Laboratories (North Liberty, IA, USA) and contained cation-adjusted Mueller-Hinton broth for comparator agents.
- Susceptibility testing for cefiderocol used broth microdilution panels containing iron-depleted media per CLSI guidelines.
- Quality assurance was performed by sterility checks, colony counts, and testing CLSI-recommended quality control reference strains.
- MIC interpretations were performed using CLSI breakpoints for comparators, and cefiderocol used the FDA/CLSI (≤4/8/≥16 mg/L for susceptible, intermediate, and resistant) and EUCAST (≤2/>2 mg/L for susceptible and resistant) breakpoints.
- Enterobacterales displaying MIC values ≥2 mg/L for imipenem (excluded for *P. mirabilis, P. penneri*, and indole-positive Proteeae) or meropenem were subjected to genome sequencing and screening of β-lactamase genes.

Screening of β-lactamase genes

- Selected isolates had total genomic DNA extracted by the fully automated Thermo Scientific™ KingFisher™ Flex Magnetic Particle Processor (Cleveland, OH, USA), which was used as input material for library construction.
- DNA libraries were prepared using the Nextera™ or Illumina DNA Prep™ library construction protocol (Illumina, San Diego, CA, USA) following the manufacturer's instructions and were sequenced on MiSeq or NextSeq Sequencer platforms at JMI Laboratories.
- FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.15.3. An in-house software was applied to align the assembled sequences against a comprehensive in-house database containing known β-lactamase genes.

Results

- A total of 2.9% (943/32,053) of all Enterobacterales isolates were nonsusceptible to imipenem and/or meropenem, of which 1.4% (207/15,149) and 4.4% (736/16,904) originated from US and European sites, respectively.
- Carbapenemase genes were detected in 77.0% (726/943) of all carbapenem-nonsusceptible Enterobacterales (Figure 1).
 - Carbapenem-nonsusceptible Enterobacterales carrying only class B MBL genes were detected in 18.0% (170/943) of the isolates and were included in this study (Figure 1).
- The activity of cefiderocol and comparator agents is shown in Table 1. For additional information, Poster P1360 describes the activity of cefiderocol and comparators against Enterobacterales carrying class A and class D carbapenemases collected in US hospitals.
- Cefiderocol (82.6–96.6% susceptible) had equivalent MIC_{90} of 4 mg/L against all carbapenem-nonsusceptible Enterobacterales isolates, including against the US and European subsets.
 - Other agents had lower susceptibilities (≤86.5%), including meropenem-vaborbactam (83.1% susceptible) and ceftazidime-avibactam (86.5% susceptible) against US isolates.
 - Even lower susceptibilities (58.4–70.2% susceptible) were noted for these two combinations against Enterobacterales collected in Europe.
- Cefiderocol had $MIC_{50/90}$ of 2/4 mg/L against the overall collection of isolates carrying MBL genes and inhibited 90.6% of these isolates at the CLSI breakpoint for susceptibility.
 - In contrast, other comparator agents had off-scale MIC_{90} values (i.e. >8 mg/L) and susceptibilities <15%, except for colistin (70.6% susceptible).
- MIC_{50} values of 2 mg/L and MIC_{90} of 4–8 mg/L were obtained for cefiderocol against subsets of isolates carrying bla_{NDM} alleles, inhibiting 84.2–91.4% of these subsets at the CLSI breakpoint for susceptibility.
 - Comparator agents showed limited activity against isolates carrying bla_{NDM} genes, except for colistin (94.7% susceptible) tested against bla_{NDM-5}-carrying isolates.
- Cefiderocol inhibited all isolates carrying bla_{VIM-1} at the CLSI breakpoint for susceptibility, where colistin was the only comparator showing elevated susceptibility (i.e. 91.7%) against this subset.

Figure 1. Distribution of carbapenemase genes[a] detected among carbapenem-nonsusceptible Enterobacterales collected from the US and European countries, including Turkey and Israel

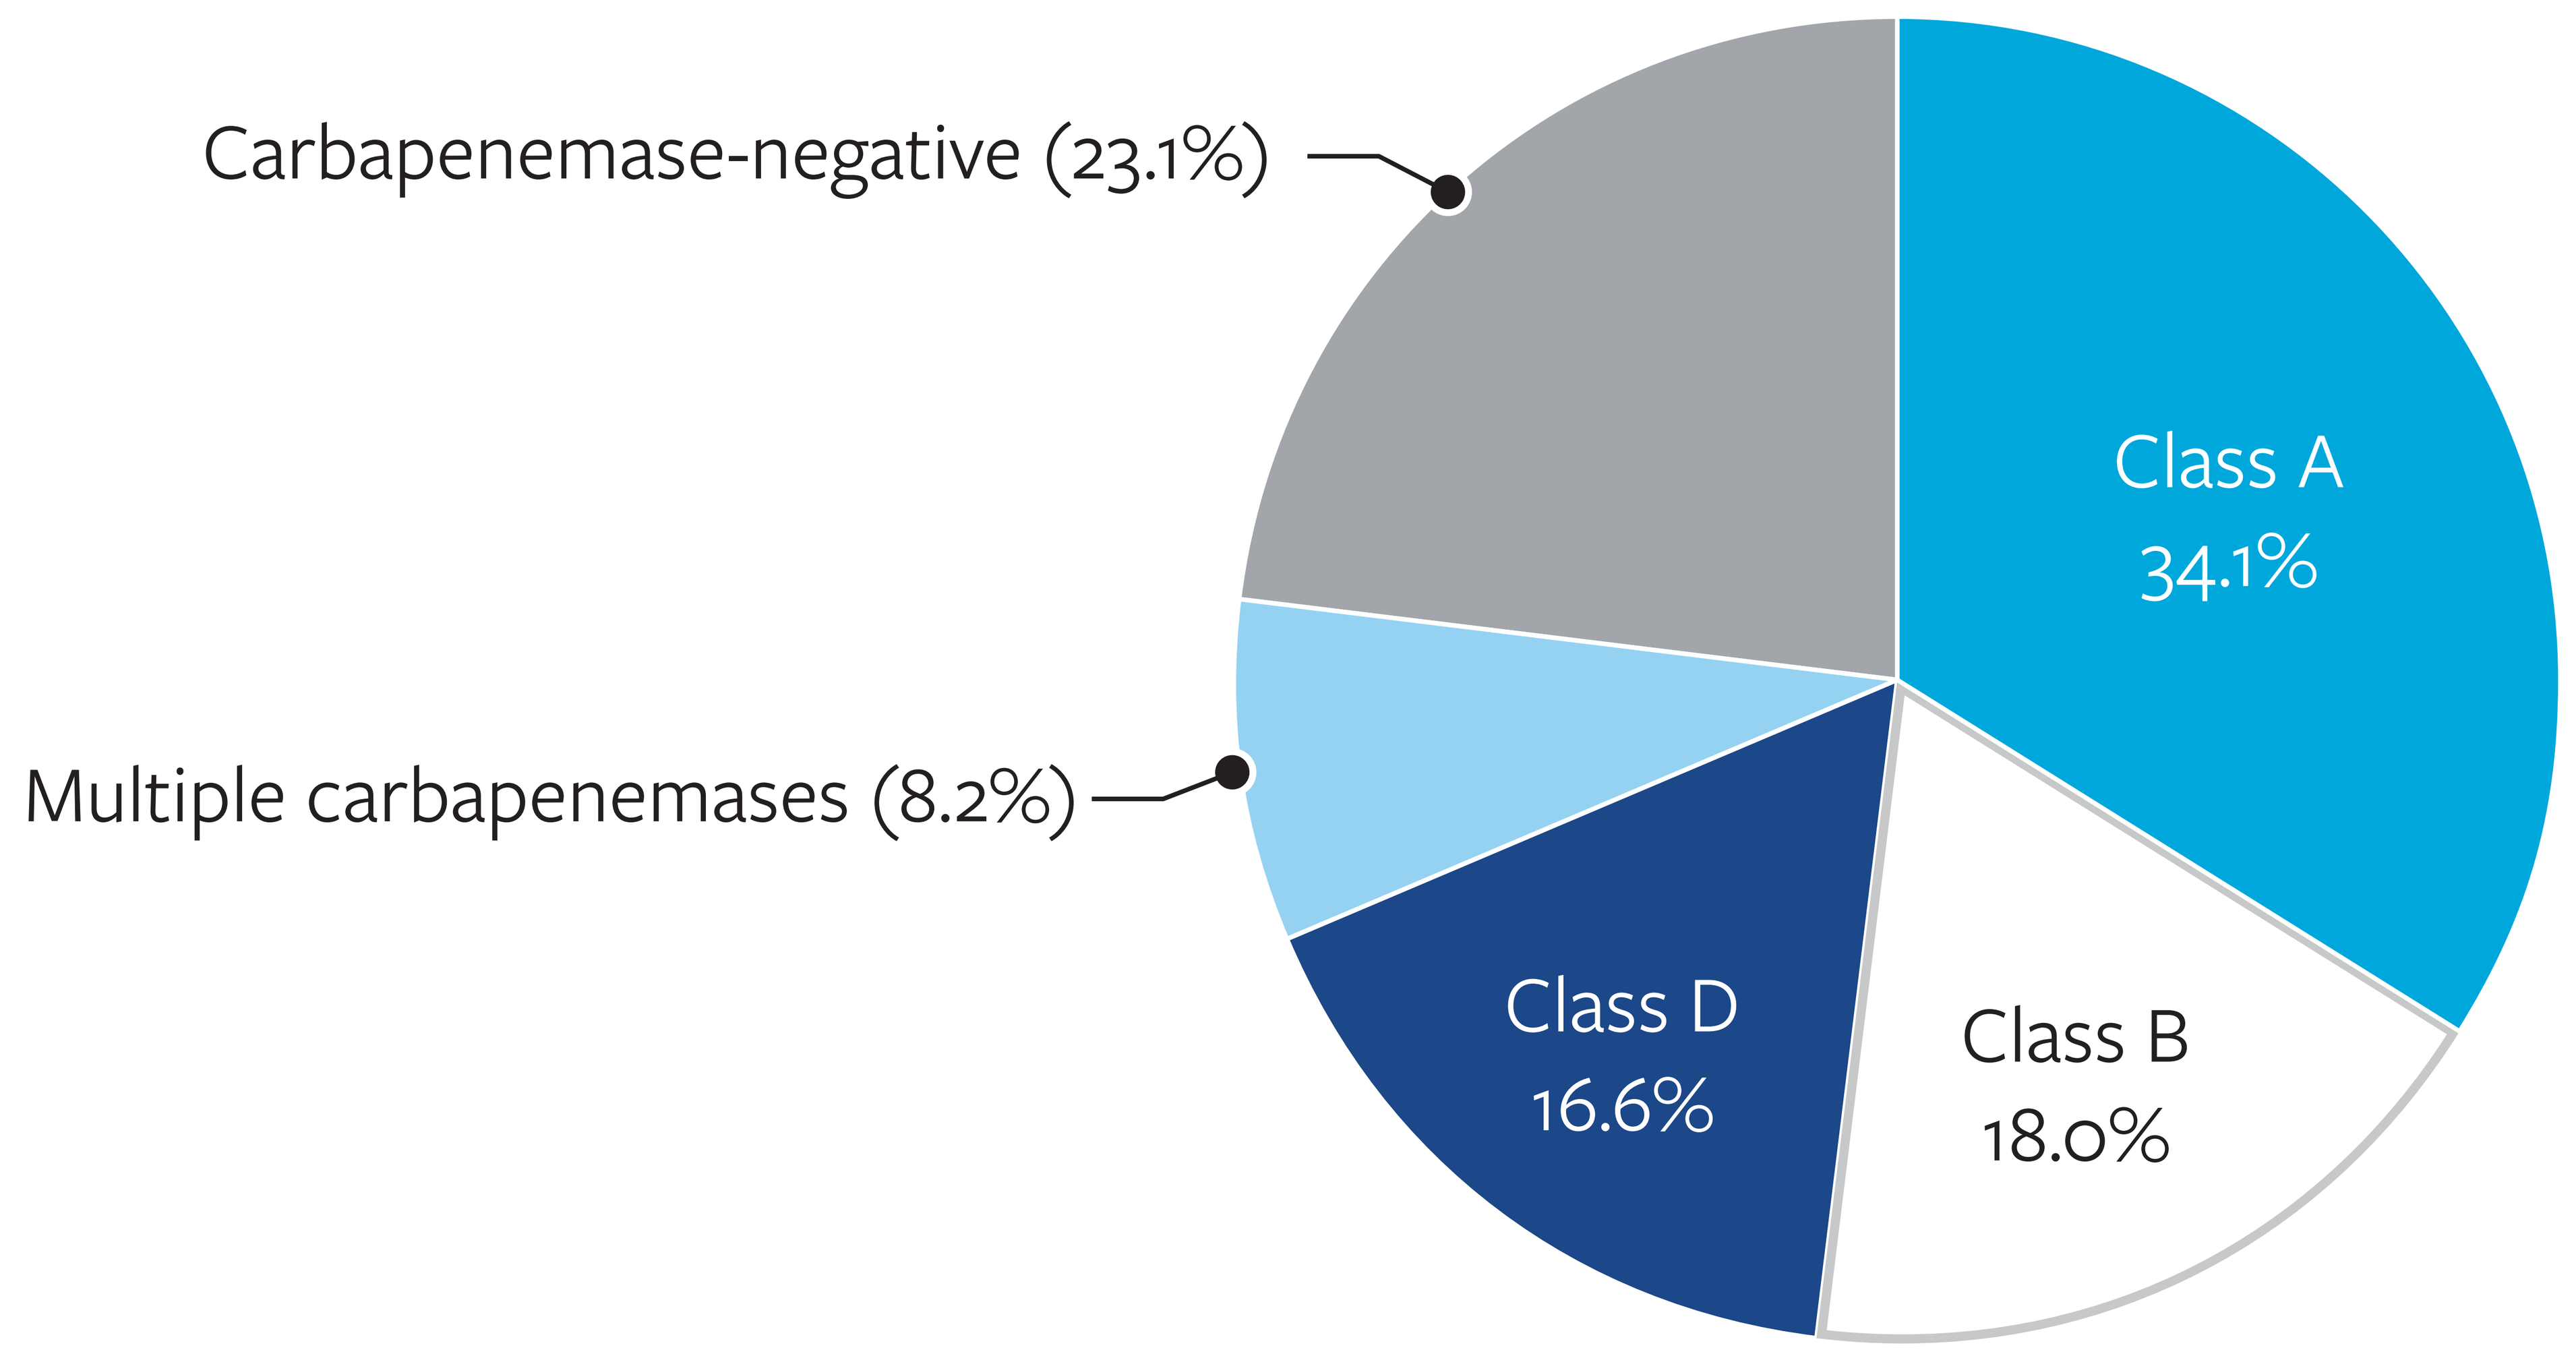
Carbapenemase-negative (23.1%)
Class A
34.1%
Multiple carbapenemases (8.2%)
Class D
16.6%
Class B
18.0%

[a] Includes the class A bla_{GES-5} (1), bla_{IMI-4} (1), bla_{KPC-2} (124), bla_{KPC-3} (190), bla_{KPC-4} (2), bla_{KPC-29} (1), bla_{SME-2} (2), and bla_{SME-4} (1); class B bla_{IMP-4} (1), bla_{NDM-1} (116), bla_{NDM-5} (19), bla_{NDM-6} (1), bla_{NDM-7} (4), bla_{NDM-9} (2), bla_{NDM-19} (1), bla_{VIM-1} (24), bla_{VIM-4} (1), and bla_{VIM-78} (1); class D $bla_{OXA-181}$ (10), $bla_{OXA-232}$ (57), $bla_{OXA-244}$ (5), and bla_{OXA-48} (85); and 77 isolates with multiple carbapenemase genes.

Table 1. Activity of cefiderocol and comparator agents tested against carbapenem-nonsusceptible Enterobacterales and resistant subsets collected from US and European countries, including Turkey and Israel

Phenotype/genotype[a] (No.)	MIC_{50}/MIC_{90} in mg/L (% susceptible)[b]					
	FDC	IMR	MEV	CZA	MER	COL
Carbapenem-nonsusceptible (943)	1/4 (95.1/84.0)	1/>8 (56.5)	1/>8 (63.8)	1/>32 (73.8)	16/>32 (16.1)	0.25/>8 (70.9)
US (207)	0.5/4 (96.6/82.6)	0.25/>8 (75.8)	0.12/>8 (83.1)	1/>32 (86.5)	4/>32 (24.2)	0.25/>8 (78.6)
Europe (736)	1/4 (94.7/82.9)	2/>8 (51.1)	2/>8 (58.4)	2/>32 (70.2)	32/>32 (13.9)	0.25/>8 (68.7)
MBL-positive[c] (170)	2/4 (90.6/64.1)	>8/>8 (0.6)	>8/>8 (14.1)	>32/>32 (2.4)	32/>32 (5.9)	0.25/>8 (70.6)
bla_{NDM} (143)	2/8 (88.8/60.8)	>8/>8 (0.7)	>8/>8 (4.2)	>32/>32 (0.0)	>32/>32 (0.7)	0.25/>8 (67.8)
bla_{NDM-1} (116)	2/4 (91.4/62.1)	>8/>8 (0.9)	>8/>8 (4.3)	>32/>32 (0.0)	32/>32 (0.9)	0.25/>8 (64.7)
bla_{NDM-5} (19)	2/8 (84.2/63.2)	>8/>8 (0.0)	>8/>8 (5.3)	>32/>32 (0.0)	>32/>32 (0.0)	0.25/0.5 (94.7)
bla_{VIM-1} (24)	1/4 (100/79.2)	4/8 (0.0)	2/>8 (66.7)	>32/>32 (8.3)	2/32 (33.3)	0.25/0.25 (91.7)

Abbreviations: FDC, cefiderocol; IMR, imipenem-relebactam; MEV, meropenem-vaborbactam; CZA, ceftazidime-avibactam; MER, meropenem; COL, colistin.
[a] Carbapenem-nonsusceptible isolates were those nonsusceptible to imipenem (excluded for *P. mirabilis, P. penneri*, and indole-positive Proteeae) and/or meropenem based on CLSI criteria (MIC values ≥2 mg/L).
[b] Cefiderocol MIC results were interpreted according to the CLSI (CLSI and the FDA criteria are the same) and EUCAST criteria. MIC values for comparator agents were interpreted based on the CLSI criteria, except for colistin where the EUCAST susceptible breakpoint was applied.
[c] Includes the class B bla_{IMP-4} (1), bla_{NDM-1} (116), bla_{NDM-5} (19), bla_{NDM-6} (1), bla_{NDM-7} (4), bla_{NDM-9} (2), bla_{NDM-19} (1), bla_{VIM-1} (24), bla_{VIM-4} (1), and bla_{VIM-78} (1) genes.

Conclusions

- Cefiderocol showed potent activity against carbapenem-nonsusceptible Enterobacterales and the subsets carrying MBL genes.
 - This potent cefiderocol activity was presented against a particular subset of resistant Enterobacterales, for which antibiotic treatment options are limited.
- Cefiderocol should be considered as an important option for the treatment of infections caused by these resistant organisms.

Acknowledgements

This research and poster presentation were sponsored by Shionogi & Co., LTD.

References

1. Clinical and Laboratory Standards Institute. 2018. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. *M07 11th Edition*. Wayne, PA, USA.

2. Clinical and Laboratory Standards Institute. 2024. Performance standards for antimicrobial susceptibility testing. *M100 34th Edition*. Wayne, PA, USA.

3. FDA Susceptibility Test Interpretive Criteria: https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed April 2024.

4. Mendes RE, Jones RN, Woosley LN, Cattoir V, Castanheira M. 2019. Application of next-generation sequencing for characterization of surveillance and clinical trial isolates: Analysis of the distribution of β-lactamase resistance genes and lineage background in the United States. *Open Forum Infect Dis* 6: S69–S78.

5. Ong'uti S, Czech M, Robilotti E, Holubar M. 2021. Cefiderocol: A new cephalosporin stratagem against multidrug resistant Gram-negative bacteria. *Clin Infect Dis*. 74: 1303–1312.

6. Potter RF, D'Souza AW, Dantas G. 2016. The rapid spread of carbapenem-resistant Enterobacteriaceae. *Drug Resist Updat*. 29: 30–46.

7. Yuan P-B, Dai L-T, Zhang Q-K, Zhong Y-X, Liu W-T, Yang L, Chen D-Q. 2024. Global emergence of double and multi-carbapenemase producing organisms: Epidemiology, clinical significance, and evolutionary benefits on antimicrobial resistance and virulence. *Microbiol Spectr*. 12: e0000824.

Contact

element

Rodrigo E. Mendes, Ph.D., FIDSA
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo.mendes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2024_24-SHI-02_A1_MBL_US_EU.pdf

Charges may apply. No personal information is stored.

IDWeek 2024, October 16–19, 2024, Los Angeles, California