IDWEEK 2024 | Poster #P1364

Cefiderocol Activity against *Pseudomonas aeruginosa* Clinical Isolates Carrying Metallo-β-Lactamase Genes in United States and European Hospitals (2020–2023)

RE Mendes, D Beekman, M Karr, JH Kimbrough, HS Sader, M Castanheira
Element Iowa City (JMI Laboratories), North Liberty, IA, USA

Introduction

- *Pseudomonas aeruginosa* isolates possess various intrinsic treatment-limiting resistance mechanisms, leading to decreased antibiotic permeability.
 - In addition, isolates may acquire β-lactamase genes, such as those encoding class A carbapenemases and especially class B metallo-β-lactamases (MBL), further decreasing susceptibility to numerous β-lactams.
 - Dissemination of MBL genes is of particular concern within European hospitals and other geographic regions.
- Cefiderocol is approved by the US Food and Drug Administration (FDA) for the treatment of complicated urinary tract infections, including pyelonephritis, as well as hospital-acquired bacterial pneumonia and ventilator-associated bacterial pneumonia.
 - Cefiderocol is also approved in Europe for the treatment of infections in adult patients caused by aerobic Gram-negative organisms with limited treatment options.
- Cefiderocol is a siderophore cephalosporin with broad activity against Gram-negative bacteria, including multidrug-resistant (MDR) organisms like carbapenem-resistant Enterobacterales (CRE), carbapenem-resistant *Pseudomonas aeruginosa*, and *Acinetobacter baumannii*.
- The antibacterial activity of this molecule is due to its ability to achieve high periplasmic concentrations by hijacking the bacterial iron transport machinery, which increases cell entry.
 - In addition, cefiderocol remains stable to hydrolysis by serine β-lactamases (ESBLs, KPCs, and OXA-type carbapenemases) and MBLs.
- In this study, cefiderocol and comparator activities were analyzed against *P. aeruginosa* carrying MBL genes, as part of the SENTRY Antimicrobial Surveillance Program for the US and Europe during 2020–2023.

Materials and Methods

Bacterial organisms

- This study comprised a collection of 9,572 *P. aeruginosa* collected from various clinical specimens from patients hospitalized in 36 medical centers in the US and 43 sites in Europe, including Turkey and Israel, during 2020–2023. Only consecutive isolates (1 per patient infection episode) responsible for documented infections according to local criteria were included.
- Bacterial identification was confirmed by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following the Clinical and Laboratory Standards Institute (CLSI) M07 (2018) guidelines.
- Frozen-form broth microdilution panels were manufactured by JMI Laboratories (North Liberty, IA, USA) and contained cation-adjusted Mueller-Hinton broth for comparator agents.
- Susceptibility testing for cefiderocol used broth microdilution panels containing iron-depleted media per CLSI guidelines.
- Quality assurance was performed by sterility checks, colony counts, and testing CLSI-recommended quality control reference strains.
- Cefiderocol MIC results were interpreted according to the CLSI, EUCAST, and FDA criteria, whereas comparator agent MIC values were interpreted based on CLSI breakpoints, except for colistin where EUCAST criteria were used.
- Carbapenem-nonsusceptible isolates were those nonsusceptible to imipenem and/or meropenem based on CLSI criteria (MIC ≥4 mg/L), and these isolates were screened for β-lactamase genes.

Screening of β-lactamase genes

- Selected isolates had total genomic DNA extracted by the fully automated Thermo Scientific™ KingFisher™ Flex Magnetic Particle Processor (Cleveland, OH, USA), which was used as input material for library construction.
- DNA libraries were prepared using the Nextera™ or Illumina DNA Prep™ library construction protocol (Illumina, San Diego, CA, USA) following the manufacturer's instructions and were sequenced on MiSeq or NextSeq Sequencer platforms at JMI Laboratories.
- FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.15.3. An in-house software was applied to align the assembled sequences against a comprehensive in-house database containing known β-lactamase genes.

Results

- Carbapenem-nonsusceptible *P. aeruginosa* comprised 23.4% (2,236/9,572) of isolates included in the cefiderocol surveillance program.
 - Among these carbapenem-nonsusceptible *P. aeruginosa*, 22.3% (980/4,400) and 24.3% (1,256/5,172) originated from US and European sites, respectively (Tables 1 and 2).
 - A total of 5.6% (126/2,236) of carbapenem-nonsusceptible *P. aeruginosa* carried MBL, and these isolates were mostly from European hospitals (94.4%) (Table 1).
 - A small number of isolates (1.0%; 23/2,236) carried other carbapenemase genes, such as $bla_{GES\text{-}5}$ (18), $bla_{GES\text{-}6}$ (4), and $bla_{KPC\text{-}2}$ (1) alone and were not included in this analysis.
- Cefiderocol (96.0–99.4% susceptible) had MIC_{50} of 0.12 mg/L and MIC_{90} of 0.5 mg/L against the overall population of carbapenem-nonsusceptible *P. aeruginosa*, as well as the US and European subsets (Table 2).
 - Comparator agents had susceptibilities (80.9–90.2%) lower than cefiderocol against carbapenem-nonsusceptible isolates, except for colistin (99.7–99.8% susceptible).
- Aztreonam-avibactam had MIC_{50} values of 8–16 mg/L and MIC_{90} of >16 mg/L when tested against all carbapenem-nonsusceptible *P. aeruginosa*, and the US and European subsets (Table 2).
- Cefiderocol (85.0–96.0% susceptible) showed activity against the MBL-carrying *P. aeruginosa* subset with an MIC_{50} value of 0.25 mg/L and MIC_{90} of 2 mg/L (Table 2).
 - In contrast, comparator agents had off-scale MIC_{90} (i.e. >8 mg/L), except for colistin ($MIC_{50/90}$, 0.5/1 mg/L), that inhibited all isolates at the EUCAST susceptible breakpoint.
- All *P. aeruginosa* carrying bla_{IMP} and bla_{VIM} were susceptible to cefiderocol ($MIC_{90/100}$, 1/2 mg/L) according to the CLSI breakpoint (Table 2).
 - Higher cefiderocol MIC results ($MIC_{50/90}$, 2/16 mg/L) were noted against bla_{NDM}-carrying *P. aeruginosa*.

Table 1. Distribution of carbapenemase genes detected among carbapenem-nonsusceptible *P. aeruginosa* collected from the US and European countries, including Turkey and Israel

Countries/Genes	Number
Belgium	**25**
VIM-2	3
Negative	22
Czech Republic	**25**
IMP-7	4
VIM-2	3
Negative	18
France	**108**
VIM-1	2
VIM-4	2
Negative	104
Germany	**176**
DIM-1	2
IMP-1	1
VIM-1, HMB-1	1
VIM-2	3
Negative	169
Greece	**59**
NDM-1	3
VIM-1	1
VIM-2	24
VIM-2, GES-5	1
VIM-4	2
Negative	28
Hungary	**45**
VIM-43	1
Negative	44
Ireland	**31**
Negative	31
Israel	**89**
IMP-13	2
VIM-4	1
Negative	86
Italy	**128**
VIM-1	1
VIM-2	13
Negative	114
Poland	**104**
VIM-1	1
VIM-2	5
VIM-4	2
Negative	96
Portugal	**21**
Negative	21
Romania	**35**
VIM-2	12
Negative	23
Slovakia	**8**
VIM-2	1
Negative	7
Slovenia	**34**
VIM-2	1
Negative	33
Spain	**143**
IMP-8	1
VIM-1	3
VIM-2	2
VIM-20	2
Negative	135
Sweden	**30**
Negative	30
Switzerland	**19**
Negative	19
Turkey	**114**
IMP-10	1
NDM-1	9
VIM-2	8
Negative	96
UK	**41**
Negative	41
USA	**978**
IMP-1	1
IMP-13	1
NDM-1	2
VIM-2	4
Negative	970
Total	**2,213[a]**

[a] A small number of isolates (1.0%; 23/2,236) carried $bla_{GES\text{-}5}$ (18), $bla_{GES\text{-}6}$ (4), and $bla_{KPC\text{-}2}$ (1) alone and were not included in this analysis.

Table 2. Activity of cefiderocol and comparator agents tested against carbapenem-nonsusceptible *P. aeruginosa* and MBL-carrying subsets collected from US and European countries, including Turkey and Israel

Phenotype/genotype[a] (No.)	MIC_{50}/MIC_{90} in mg/L (% susceptible)[b]					
	FDC	IMR	CXT	CZA	AZA	COL
Carbapenem-nonsusceptible (2,236)	0.12/0.5 (99.2/98.6/96.3)	1/4 (85.5)	1/16 (85.0)	4/16 (85.3)	8/>16 (—)	0.5/1 (99.7)
US (980)	0.12/0.5 (99.4/98.9/96.6)	1/4 (89.5)	1/4 (90.2)	4/16 (86.9)	16/>16 (—)	0.5/1 (99.8)
Europe (1,256)	0.12/0.5 (99.0/98.3/96.0)	1/>8 (82.3)	1/>16 (80.9)	4/32 (84.0)	8/>16 (—)	0.5/1 (99.7)
MBL-positive[c] (126)	0.25/2 (96.0/94.4/85.0)	>8/>8 (2.4)	>16/>16 (0.8)	>32/>32 (4.0)	8/>16 (—)	0.5/1 (100)
bla_{IMP} (11)	0.5/1 (100/100/90.9)	>8/>8 (0.0)	>16/>16 (0.0)	>32/>32 (0.0)	16/>16 (—)	0.5/1 (100)
bla_{NDM} (14)	2/16 (64.3/57.1/14.3)	>8/>8 (0.0)	>16/>16 (0.0)	>32/>32 (0.0)	4/>16 (—)	0.5/1 (100)
bla_{VIM} (99)	0.12/1 (100/99.0/93.9)	>8/>8 (1.0)	>16/>16 (1.0)	32/>32 (5.1)	8/>16 (—)	0.5/1 (100)

Abbreviations: FDC, cefiderocol; IMR, imipenem-relebactam; CXT, ceftolozane-tazobactam; CZA, ceftazidime-avibactam; AZA, aztreonam-avibactam; COL, colistin.
[a] Carbapenem-nonsusceptible isolates were those nonsusceptible to imipenem and/or meropenem based on CLSI criteria (MIC values ≥4 mg/L).
[b] Cefiderocol MIC results were interpreted according to the CLSI/EUCAST/FDA criteria for susceptibility (≤4 mg/L, ≤2 mg/L, and ≤1 mg/L, respectively). MIC values for comparator agents were interpreted based on the CLSI criteria, except for colistin where the EUCAST susceptible breakpoint was applied.
[c] Includes the class B $bla_{DIM\text{-}1}$ (2), $bla_{IMP\text{-}1}$ (2), $bla_{IMP\text{-}7}$ (4), $bla_{IMP\text{-}8}$ (1), $bla_{IMP\text{-}10}$ (1), $bla_{IMP\text{-}13}$ (3), $bla_{NDM\text{-}1}$ (14), $bla_{VIM\text{-}1}$ (8), $bla_{VIM\text{-}1}$ and $bla_{HMB\text{-}1}$ (1), $bla_{VIM\text{-}2}$ (80), $bla_{VIM\text{-}4}$ (7), $bla_{VIM\text{-}20}$ (2), and $bla_{VIM\text{-}43}$ (1) genes.

Conclusions

- Cefiderocol showed potent activity against carbapenem-nonsusceptible *P. aeruginosa* clinical isolates from US and European hospitals.
- This activity included against isolates carrying MBL genes, whereas newly launched β-lactam-β-lactamase inhibitor combinations didn't show activity.
- Cefiderocol should be considered as an important option for the treatment of infections caused by these resistant subsets for which antibiotic treatment options are limited.

Acknowledgements

This research and poster presentation were sponsored by Shionogi & Co., LTD.

References

1. Clinical and Laboratory Standards Institute. 2018. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. *M07 11th Edition*. Wayne, PA, USA.

2. Clinical and Laboratory Standards Institute. 2024. Performance standards for antimicrobial susceptibility testing. *M100 34th Edition*. Wayne, PA, USA.

3. FDA Susceptibility Test Interpretive Criteria: https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed April 2024.

4. Mendes RE, Jones RN, Woosley LN, Cattoir V, Castanheira M. 2019. Application of next-generation sequencing for characterization of surveillance and clinical trial isolates: Analysis of the distribution of β-lactamase resistance genes and lineage background in the United States. *Open Forum Infect Dis* 6: S69–S78.

5. Ong'uti S, Czech M, Robilotti E, Holubar M. 2021. Cefiderocol: A new cephalosporin stratagem against multidrug resistant Gram-negative bacteria. *Clin Infect Dis*. 74: 1303–1312.

Contact

element

SCAN ME

Rodrigo E. Mendes, Ph.D., FIDSA
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo.mendes@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2024_24-SHI-02_A2_MBL_PSA_US_EU.pdf

Charges may apply. No personal information is stored.

IDWeek 2024, October 16–19, 2024, Los Angeles, California