IDWEEK 2024 | Poster #P1362

Cefiderocol Activity against Carbapenem-Resistant *Acinetobacter baumannii-calcoaceticus* Complex, Including Molecularly Characterized Clinical Isolates, Causing Infections in United States Hospitals (2020–2023)

RE Mendes, A Scullin, JH Kimbrough, M Karr, JM Maher, M Castanheira

Element Iowa City (JMI Laboratories), North Liberty, IA, USA

Introduction

- Multidrug-resistant (MDR) *Acinetobacter baumannii-calcoaceticus* complex has gained attention as an important clinical challenge in the last decades, due to its ability to develop resistance to front-line antibiotics.
- Cefiderocol is approved by the US Food and Drug Administration (FDA) for the treatment of complicated urinary tract infections (UTI), including pyelonephritis, as well as hospital-acquired bacterial pneumonia and ventilator-associated bacterial pneumonia.
- Cefiderocol is a siderophore cephalosporin with broad activity against Gram-negative bacteria, including multidrug-resistant (MDR) organisms like carbapenem-resistant Enterobacterales (CRE), carbapenem-resistant *Pseudomonas aeruginosa*, and *A. baumannii-calcoaceticus* complex.
- In this study, the activities of cefiderocol and comparator agents were evaluated against *A. baumannii-calcoaceticus* complex causing infections in US hospitals, including molecularly characterized resistant subsets, as part of the SENTRY Antimicrobial Surveillance Program during 2020–2023.

Materials and Methods

Bacterial organisms

- This study comprised a collection of 1,786 *Acinetobacter* spp. cultured from various clinical specimens in patients hospitalized in 72 medical centers in all 9 US Census Divisions during 2020–2023.
- Only consecutive isolates (1 per patient infection episode) responsible for documented infections according to local criteria were included.
- Bacterial identification was confirmed by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following the Clinical and Laboratory Standards Institute (CLSI) M07 (2018) guidelines.
- Frozen-form broth microdilution panels were manufactured by Element Iowa City (JMI Laboratories) (North Liberty, IA, USA) and contained cation-adjusted Mueller-Hinton broth for comparator agents.
- Susceptibility testing for cefiderocol used broth microdilution panels containing iron-depleted media per CLSI guidelines.
- Quality assurance was performed by sterility checks, colony counts, and testing CLSI-recommended quality control reference strains.
- Cefiderocol MIC results were interpreted according to the CLSI/FDA criteria, whereas comparator agent MIC values were interpreted based on the FDA susceptible breakpoint for imipenem-relebactam, EUCAST susceptible breakpoint for colistin, and CLSI criteria for all other agents.
- Carbapenem-nonsusceptible *A. baumannii-calcoaceticus* complex isolates were those nonsusceptible to imipenem and/or meropenem based on CLSI criteria (MIC, ≥4 mg/L). These isolates were subjected to genome sequencing for screening of β-lactamase genes.

Screening of β-lactamase genes

- Selected isolates had total genomic DNA extracted by the fully automated Thermo Scientific™ KingFisher™ Flex Magnetic Particle Processor (Cleveland, OH, USA), which was used as input material for library construction.
- DNA libraries were prepared using the Nextera™ or Illumina DNA Prep™ library construction protocol (Illumina, San Diego, CA, USA) following the manufacturer's instructions and were sequenced on MiSeq or NextSeq Sequencer platforms at JMI Laboratories.
- FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.15.3. An in-house software was applied to align the assembled sequences against a comprehensive in-house database containing known β-lactamase genes.

Results

- Among *Acinetobacter* spp. clinical isolates included here, the majority were responsible for pneumonia (39%) and skin and skin-structure infections (25%), followed by bloodstream (15%) and urinary tract infections (13%) (Figure 1).
- *A. baumannii-calcoaceticus* complex comprised 80.5% (1,438/1,786) of *Acinetobacter* spp. isolates included, followed by *A. ursingii* (5.1%; 91/1,786), *A. radioresistens* (3.8%; 68/1,786), *A. bereziniae* (2.2%; 40/1,786), and *A. lwoffii* (2.2%; 39/1,786) (Table 1).
 - Other *Acinetobacter* spp. isolates were represented by 20 isolates or less.
- A total of 20.0% (411/1,786) of *Acinetobacter* spp. were carbapenem-nonsusceptible. All but 6 (98.5%; 405/411) were identified as *A. baumannii-calcoaceticus* complex.
- Carbapenemase genes were detected in 86.9% (352/405) of carbapenem-nonsusceptible *A. baumannii-calcoaceticus* complex (Figure 2 and Table 1).
 - bla_{OXA-23} (60.0%; 211/352) prevailed, followed by bla_{OXA-24} (34.9%; 123/352).
- Cefiderocol (93.6–98.8% susceptible) had MIC_{50} of 0.12 mg/L and MIC_{90} of 1 mg/L against all *Acinetobacter* spp. and the *A. baumannii-calcoaceticus* complex subset (Table 1).
 - Comparators had limited activity against these isolates (66.5–77.1% susceptible), except for colistin (95.7–96.2% susceptible).
- Cefiderocol (97.0–97.7% susceptible by CLSI) had MIC_{50} of 0.25 mg/L and MIC_{90} of 2 mg/L against carbapenem-nonsusceptible *A. baumannii-calcoaceticus* complex and those carrying carbapenemases (Table 1).
 - Only colistin was also *in vitro* active against these two resistant subsets.
- Cefiderocol (97.2–99.2% susceptible by CLSI) MIC_{90} of 2 mg/L and 1 mg/L were obtained against *A. baumannii-calcoaceticus* complex carrying bla_{OXA-23} and bla_{OXA-24}, respectively (Table 1).
- Isolates carrying other carbapenemases were inhibited by cefiderocol MIC of ≤2 mg/L, except for 1 strain with bla_{OXA-23} and bla_{NDM-1} (MIC, 8 mg/L) (Table 1).

Figure 1. Distribution of infection types[a] caused by *Acinetobacter* spp.

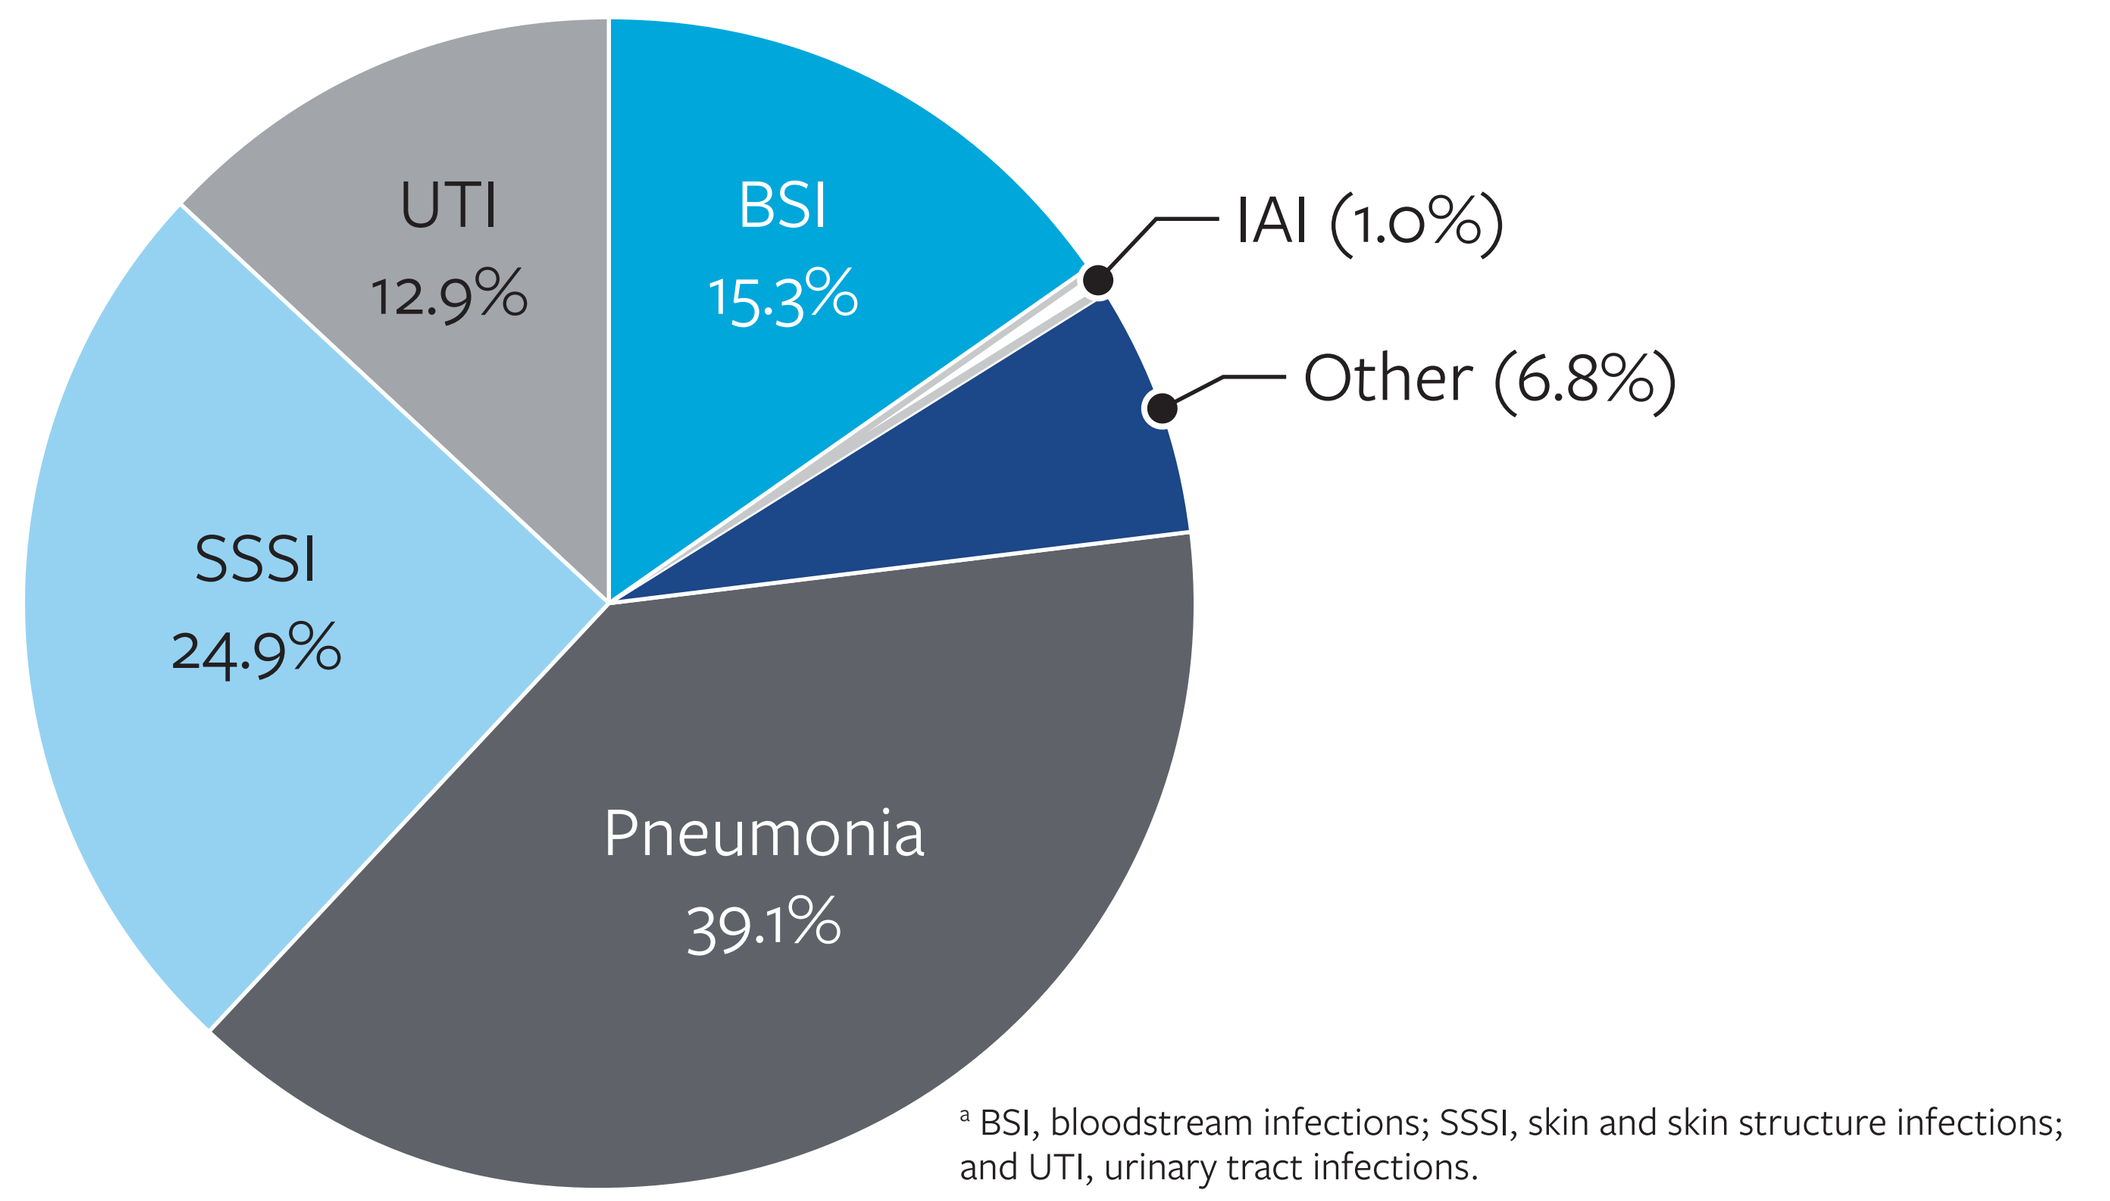

UTI
12.9%
BSI
15.3%
IAI (1.0%)
Other (6.8%)
SSSI
24.9%
Pneumonia
39.1%

[a] BSI, bloodstream infections; SSSI, skin and skin structure infections; and UTI, urinary tract infections.

Figure 2. Distribution of carbapenemase genes detected in carbapenem-nonsusceptible *A. baumannii-calcoaceticus* group

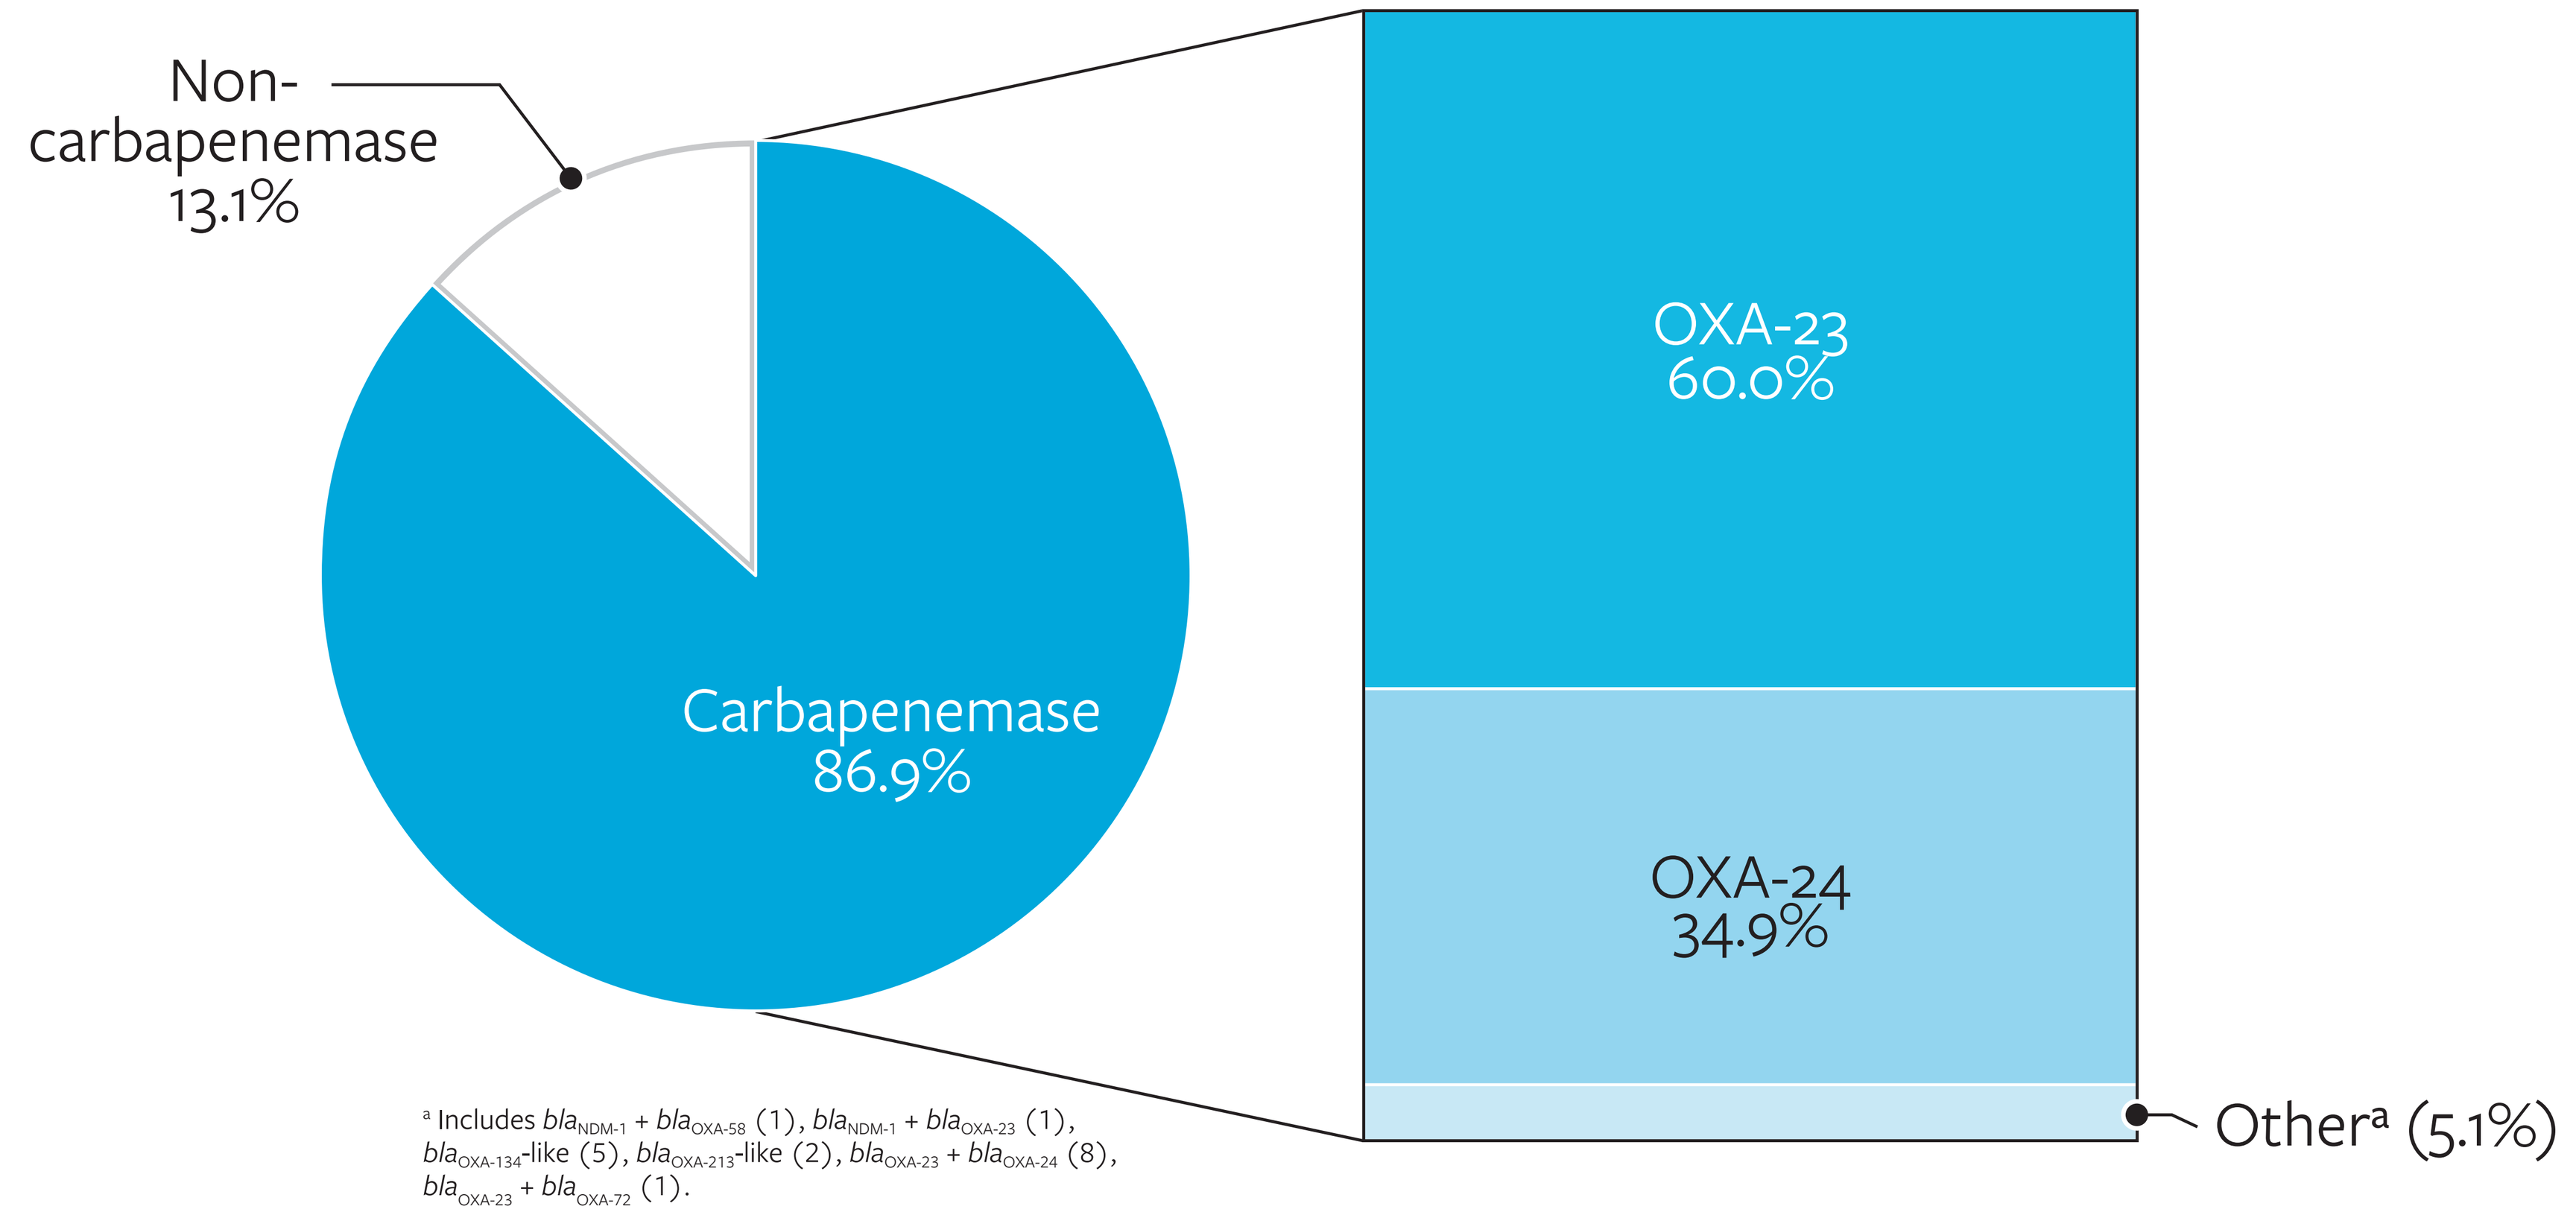

Non-carbapenemase
13.1%
Carbapenemase
86.9%
OXA-23
60.0%
OXA-24
34.9%
Other[a] (5.1%)

[a] Includes bla_{NDM-1} + bla_{OXA-58} (1), bla_{NDM-1} + bla_{OXA-23} (1), $bla_{OXA-134}$-like (5), $bla_{OXA-213}$-like (2), bla_{OXA-23} + bla_{OXA-24} (8), bla_{OXA-23} + bla_{OXA-72} (1).

Table 1. Activity of cefiderocol and comparator agents against *Acinetobacter* spp. and resistant subsets from the USA

Phenotype[a]/genotype (No.)	MIC_{50}/MIC_{90} in mg/L (% susceptible by CLSI/FDA criteria)[b]					
	FDC	IMR	MER	A/S	CAZ	COL
All[c] (1,786)	0.12/1 (98.8/94.2)	0.25/>8 (77.1)	0.5/>32 (75.9)	4/64 (72.9)	8/>32 (68.3)	0.5/1 (95.7)
ACB (1,438)	0.12/1 (98.5/93.6)	0.25/>8 (71.8)	0.5/>32 (70.4)	4/64 (67.5)	8/>32 (66.5)	0.5/1 (96.2)
Carbapenem-nonsusceptible[d] (405)	0.25/2 (97.0/86.9)	>8/>8 (4.2)	>32/>32 (0.2)	32/>64 (13.3)	>32/>32 (17.3)	0.5/1 (96.0)
Carbapenemase-positive (352)	0.25/2 (97.7/87.8)	>8/>8 (0.6)	>32/>32 (0.0)	32/>64 (5.7)	>32/>32 (19.3)	0.5/1 (96.0)
OXA-23 (211)	0.5/2 (97.2/85.3)	>8/>8 (0.0)	>32/>32 (0.0)	32/>64 (0.9)	>32/>32 (14.7)	0.5/1 (94.8)
OXA-24 (123)	0.25/1 (99.2/93.5)	>8/>8 (0.0)	>32/>32 (0.0)	32/>64 (11.4)	32/>32 (25.2)	0.5/0.5 (98.4)
Other[e] (18)	0.25/2 (94.4/77.8)	>8/>8 (11.1)	>32/>32 (0.0)	32/>64 (22.2)	32/>32 (33.3)	0.5/1 (94.4)
Carbapenemase-negative[f] (53)	0.25/4 (92.5/81.1)	8/>8 (28.3)	16/32 (1.9)	8/64 (64.2)	>32/>32 (3.8)	0.5/2 (96.2)

Abbreviations: ACB, *A. baumannii-calcoaceticus* species complex; FDC, cefiderocol; IMR, imipenem-relebactam; MER, meropenem; A/S, ampicillin-sulbactam; CAZ, ceftazidime; COL, colistin.
[a] Carbapenem-nonsusceptible, isolates nonsusceptible to imipenem and/or meropenem based on CLSI criteria (MIC values ≥4 mg/L).
[b] Cefiderocol MIC results were interpreted according to the CLSI/FDA criteria, whereas comparator agent MIC values were interpreted based on the FDA susceptible breakpoint for IMR, EUCAST susceptible breakpoint for colistin, and CLSI criteria for other agents.
[c] Includes *A. baumannii-calcoaceticus* species complex (1,438), *A. beijerinckii* (3), *A. bereziniae* (40), *A. courvalinii* (13), *A. dispersus* (2), *A. guillouiae* (5), *A. gyllenbergii* (1), *A. haemolyticus* (4), *A. indicus* (1), *A. johnsonii* (9), *A. junii* (20), *A. lwoffii* (39), *A. nosocomialis* (1), *A. proteolyticus* (7), *A. radioresistens* (68), *A. schindleri* (5), *A. soli* (2), *A. ursingii* (91), *A. variabilis* (15), *A. venetianus* (1), *A. vivianii* (7), and *Acinetobacter* spp. (14).
[d] All *A. baumannii-calcoaceticus* species complex.
[e] Includes bla_{NDM-1} + bla_{OXA-58} (1), bla_{NDM-1} + bla_{OXA-23} (1), $bla_{OXA-134}$-like (5), $bla_{OXA-213}$-like (2), bla_{OXA-23} + bla_{OXA-24} (8), bla_{OXA-23} + bla_{OXA-72} (1).
[f] Acquired carbapenemase genes not detected.

Conclusions

- This study demonstrates the MDR nature of *A. baumannii-calcoaceticus* complex causing infections in US hospitals.
- Cefiderocol remained as an active agent against these isolates, regardless of resistance genotype, whereas comparator agents, with the exception of colistin, showed limited *in vitro* activity.
- These *in vitro* data suggest cefiderocol as an important option for the treatment of infections caused by MDR *A. baumannii-calcoaceticus* complex, for which antibiotic treatment options are limited.

Acknowledgements

This research and poster presentation were sponsored by Shionogi & Co., LTD.

References

1. Clinical and Laboratory Standards Institute. 2018. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. *M07 11th Edition*. Wayne, PA, USA.

2. Clinical and Laboratory Standards Institute. 2024. Performance standards for antimicrobial susceptibility testing. *M100 34th Edition*. Wayne, PA, USA.

3. FDA Susceptibility Test Interpretive Criteria: https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed April 2024.

4. Karlowsky JA, Hackel MA, Takemura M, Yamano Y, Echols R, Sahm DF. 2022. *In vitro* susceptibility of Gram-negative pathogens to cefiderocol in five consecutive annual multinational SIDERO-WT Surveillance Studies, 2014 to 2019. *Antimicrob Agents Chemother*. 66: e0199021.

5. Mendes RE, Jones RN, Woosley LN, Cattoir V, Castanheira M. 2019. Application of next-generation sequencing for characterization of surveillance and clinical trial isolates: Analysis of the distribution of β-lactamase resistance genes and lineage background in the United States. *Open Forum Infect Dis* 6: S69–S78.

6. Ong'uti S, Czech M, Robilotti E, Holubar M. 2022. Cefiderocol: A new cephalosporin stratagem against multidrug resistant Gram-negative bacteria. *Clin Infect Dis*. 74: 1303–1312.

Contact

element

Rodrigo E. Mendes, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo.mendes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2024_23-SHI-09_A6_ACB_IDWeek2024.pdf

Charges may apply. No personal information is stored.

IDWeek 2024, October 16–19, 2024, Los Angeles, California