IDWEEK 2025 | Poster #P1287

In Vitro Activity of Cefiderocol against Resistant Subsets of *Pseudomonas aeruginosa* Collected from United States Hospitals during 2020–2024

RE Mendes, JM Maher, Z Kockler, JH Kimbrough, M Castanheira
Element Iowa City (JMI Laboratories), North Liberty, IA, USA

Introduction

- *Pseudomonas aeruginosa* possess various intrinsic treatment-limiting resistance mechanisms, leading to decreased antibiotic permeability.
 - Isolates may also acquire β-lactamase genes, such as those encoding class A carbapenemases and especially class B metallo-β-lactamases further decreasing susceptibility to numerous β-lactams.
- Cefiderocol is approved by the US Food and Drug Administration (FDA) for the treatment of complicated urinary tract infections, including pyelonephritis, as well as hospital-acquired bacterial pneumonia and ventilator-associated bacterial pneumonia.
- Cefiderocol is a siderophore cephalosporin with broad activity against Gram-negative bacteria, including multidrug-resistant (MDR) organisms like carbapenem-resistant Enterobacterales (CRE), carbapenem-resistant *P. aeruginosa*, *Acinetobacter baumannii*, and *Stenotrophomonas maltophilia*.
- In this study, the activities of cefiderocol and comparator agents were evaluated against *P. aeruginosa* causing infections in US hospitals, including resistant subsets, as part of the SENTRY Antimicrobial Surveillance Program during 2020–2024.

Materials and Methods

Bacterial organisms

- This study comprised a collection of 5,478 *P. aeruginosa* cultured from various clinical specimens in patients hospitalized in 38 medical centers in all 9 US Census Divisions during 2020–2024. Only consecutive isolates (1 per patient infection episode) responsible for documented infections according to local criteria were included.
- Bacterial identification was confirmed by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following the Clinical and Laboratory Standards Institute (CLSI) guidelines.
- Frozen-form broth microdilution panels were manufactured by Element Iowa City (JMI Laboratories) (North Liberty, IA, USA) and contained cation-adjusted Mueller-Hinton broth for comparator agents.
- Susceptibility testing for cefiderocol used broth microdilution panels containing iron-depleted media per CLSI guidelines.
- Quality assurance was performed by sterility checks, colony counts, and testing CLSI-recommended quality control reference strains.
- Cefiderocol MIC results were interpreted according to the CLSI and FDA criteria, whereas comparator agent MIC values were interpreted based on CLSI breakpoints.
- Carbapenem-nonsusceptible were those isolates nonsusceptible to imipenem and/or meropenem based on CLSI criteria (MIC, ≥4 mg/L). MDR was classified as nonsusceptible to ≥3 drug classes using CLSI breakpoints; extensively drug-resistant (XDR) was defined as nonsusceptible to all but 2 or fewer drug classes using CLSI breakpoints. Furthermore, difficult-to-treat resistance (DTR) was defined when nonsusceptibility (CLSI breakpoints) was detected for piperacillin-tazobactam, ceftazidime, cefepime, aztreonam, meropenem, imipenem, ciprofloxacin, and levofloxacin.

Screening of β-lactamase genes

- Selected isolates had total genomic DNA extracted by the fully automated Thermo Scientific™ KingFisher™ Flex Magnetic Particle Processor (Cleveland, OH, USA), which was used as input material for library construction.
- DNA libraries were prepared using the Nextera™ or Illumina DNA Prep™ library construction protocol (Illumina, San Diego, CA, USA) following the manufacturer's instructions and were sequenced on MiSeq or NextSeq Sequencer platforms at JMI Laboratories.
- FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.15.3. An in-house software was applied to align the assembled sequences against a comprehensive in-house database containing known β-lactamase genes.

Results

- A total of 14.7% (805/5,478), 3.5% (190/5,478), and 2.1% (116/5,478) *P. aeruginosa* isolates were MDR, DTR, and XDR, respectively (Table 1).
 - In addition, 22.6% (1,238/5,478) *P. aeruginosa* were carbapenem-nonsusceptible, and all but 13 isolates (98.9%; 1,225/1,238) were carbapenemase-negative.
 - Carbapenemase genes included bla_{GES-5} (2), bla_{IMP-1} (1), bla_{IMP-13} (1), bla_{IMP-83} (1), bla_{KPC-2} (1), bla_{NDM-1} (2), bla_{VIM-1} (1), and bla_{VIM-2} (4) (Table 1).
 - Most carbapenem-nonsusceptible *P. aeruginosa* originated from pneumonia patients (67%), with smaller percentages from skin and skin structure infections (14.5%), bloodstream infections (7.5%), and urinary tract infections (7.9%) (Figure 1).
- Cefiderocol and β-lactam/β-lactamase inhibitor (BL/BLI) combinations showed susceptibilities of >95% against all *P. aeruginosa*, except for piperacillin-tazobactam (79.4% susceptible) (Table 2).
- Cefiderocol (95.5–99.1% susceptible) showed MIC_{50} of 0.12 mg/L and MIC_{90} of 0.5 mg/L against MDR isolates.
- Cefiderocol also inhibited 98.4% and 96.6% of DTR ($MIC_{50/90}$, 0.12/1 mg/L) and XDR ($MIC_{50/90}$, 0.12/2 mg/L) *P. aeruginosa* isolates at the CLSI breakpoint for susceptibility, respectively (Tables 1 and 2).
 - BL/BLI combinations showed various lower degrees of susceptibilities (≤85.1% susceptible) against the MDR, DTR, and XDR subsets (Table 2).
- Cefiderocol (96.8–99.4% susceptible) had the lowest MIC results obtained against carbapenem-nonsusceptible *P. aeruginosa* with an MIC_{90} ≥4-fold lower than comparator agents (Table 2).
 - Imipenem-relebactam, ceftazidime-avibactam, and ceftolozane-tazobactam showed susceptibilities of 87.1–90.1% against carbapenem-nonsusceptible *P. aeruginosa* (Table 2).
 - Piperacillin-tazobactam inhibited 49.3% of carbapenem-nonsusceptible isolates at the CLSI breakpoint for susceptibility (Table 2).
- Cefiderocol (100% susceptible) inhibited all *P. aeruginosa* carrying carbapenemase genes at ≤2 mg/L, which is under the CLSI breakpoint for susceptibility of ≤4 mg/L (Table 1).
 - Other comparators showed susceptibilities of <25% against *P. aeruginosa* carrying carbapenemase genes.
- Cefiderocol ($MIC_{50/90}$, 0.12/0.5 mg/L; 97.0–99.4% susceptible), imipenem-relebactam ($MIC_{50/90}$, 1/2 mg/L; 90.8% susceptible), and ceftolozane-tazobactam ($MIC_{50/90}$, 1/4 mg/L; 91.0% susceptible) were the most active agents against carbapenem-nonsusceptible *P. aeruginosa* without carbapenemase genes, followed by ceftazidime-avibactam (87.8% susceptible) (Table 2).

Figure 1. Distribution of infection types[a] caused by carbapenem-nonsusceptible *P. aeruginosa*

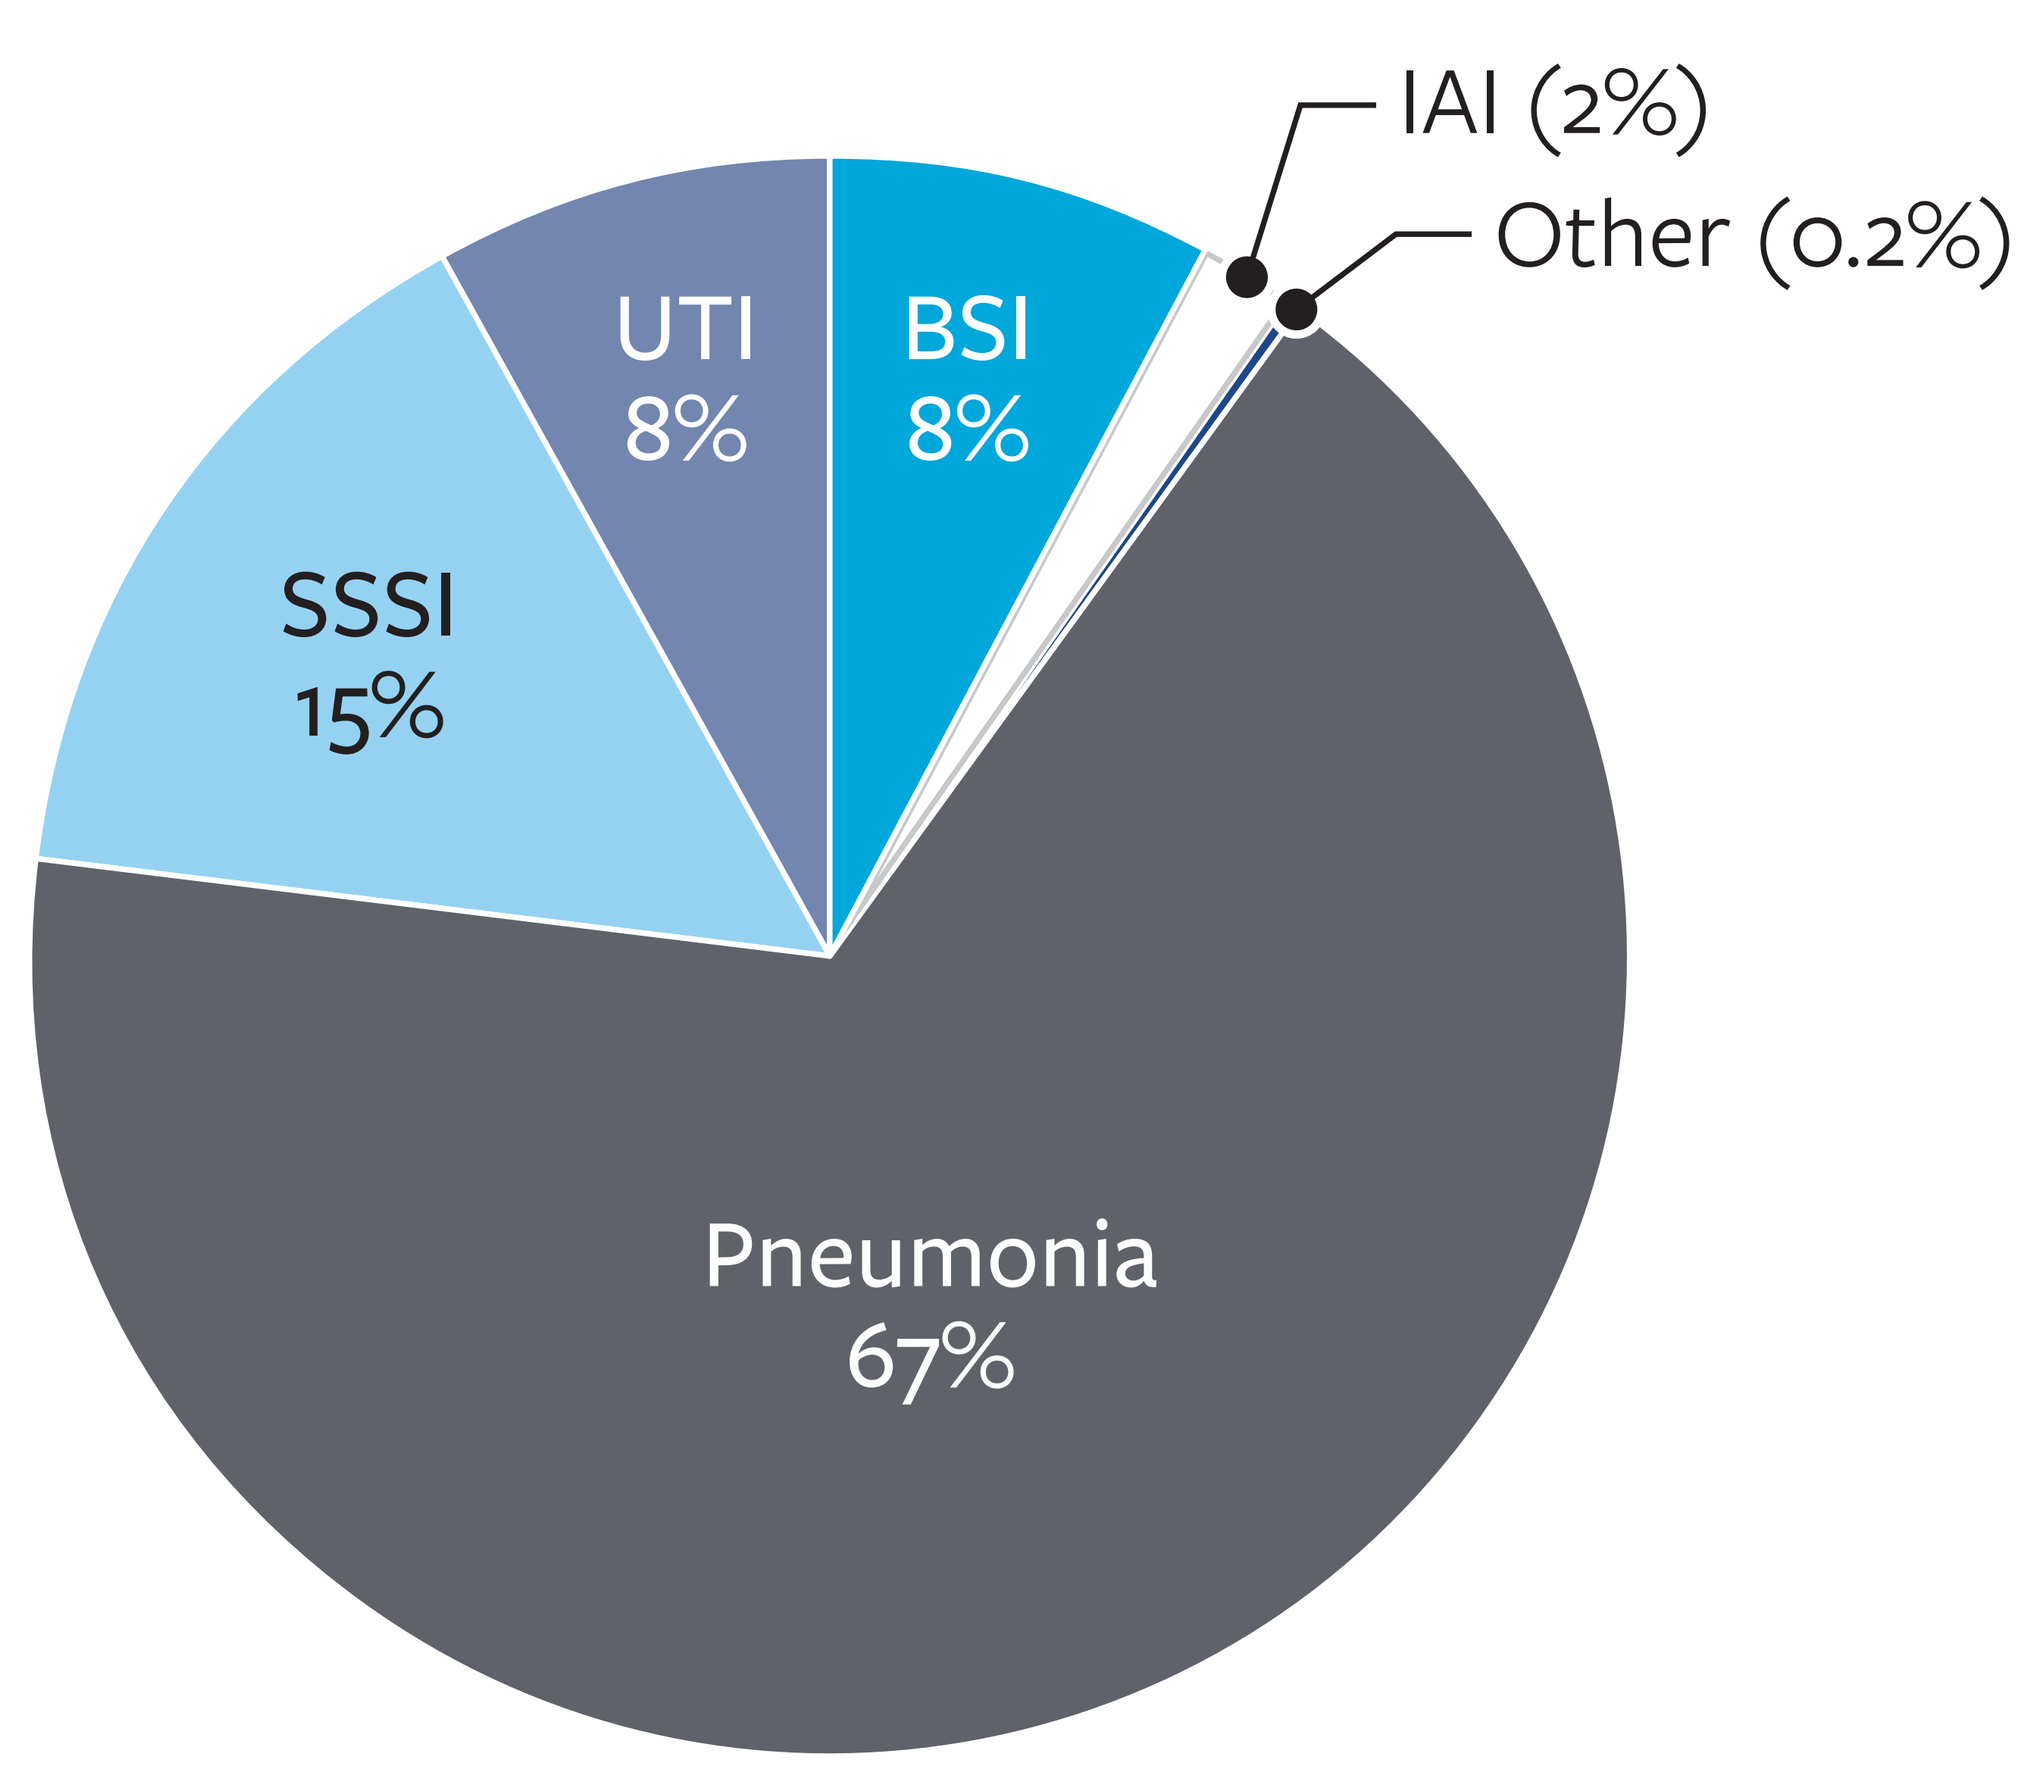
IAI (2%)
Other (0.2%)
UTI 8%
BSI 8%
SSSI 15%
Pneumonia 67%

[a] BSI, bloodstream infections; IAI, intra-abdominal infections; SSSI, skin and skin structure infections; and UTI, urinary tract infections.

Table 1. Frequency distribution of cefiderocol against *P. aeruginosa* and resistant subsets from USA hospitals

Phenotype/genotype[a] (No. tested)	Number and cumulative % of isolates inhibited at MIC (mg/L) of:											MIC	
	≤0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	>16	MIC_{50}	MIC_{90}
All (5,478)	1269 23.2	1335 47.5	1457 74.1	906 90.7	339 96.9	97 98.6	49 99.5	16 99.8	7 99.9	3 100		0.12	0.25
MDR (805)	134 16.6	141 34.2	187 57.4	179 79.6	87 90.4	41 95.5	22 98.3	7 99.1	5 99.8	2 100		0.12	0.5
DTR (190)	25 13.2	30 28.9	44 52.1	37 71.6	25 84.7	13 91.6	13 98.4	0 98.4	3 100			0.12	1
XDR (116)	16 13.8	25 35.3	23 55.2	16 69.0	15 81.9	8 88.8	9 96.6	0 96.6	4 100			0.12	2
Carbapenem-nonsusceptible (1,238)	258 20.8	256 41.5	303 66.0	224 84.1	115 93.4	43 96.8	25 98.9	7 99.4	5 99.8	2 100		0.12	0.5
Carbapenemase-positive[b] (13)		2 15.4	3 38.5	1 46.2	2 61.5	3 84.6	2 100					0.5	2
Carbapenemase-negative (1,225)	258 21.1	254 41.8	300 66.3	223 84.5	113 93.7	40 97.0	23 98.9	7 99.4	5 99.8	2 100		0.12	0.5

[a] MDR, multidrug resistance classified as non-susceptible (CLSI breakpoints) to ≥3 drug classes; DTR; difficult-to-treat resistance, defined as non-susceptible (CLSI) to piperacillin-tazobactam, ceftazidime, cefepime, aztreonam, meropenem, imipenem, ciprofloxacin, and levofloxacin; XDR, extensively drug resistant, defined as non-susceptible (CLSI) to all but 2 or fewer drug classes; carbapenem-nonsusceptible, isolates non-susceptible (MIC values ≥4 mg/L) to imipenem and/or meropenem based on CLSI criteria.
[b] Includes bla_{GES-5} (2), bla_{IMP-1} (1), bla_{IMP-13} (1), bla_{IMP-83} (1), bla_{KPC-2} (1), bla_{NDM-1} (2), bla_{VIM-1} (1), and bla_{VIM-2} (4).

Table 2. Activity of cefiderocol and β-lactam-β-lactamase inhibitor combinations against *P. aeruginosa* and resistant subsets from the USA

Phenotype/genotype[a] (No. tested)	MIC_{50}/MIC_{90} in mg/L (% susceptible by CLSI/FDA criteria)[b]					
	FDC	IMR	CZA	C/T	P/T	MER
All (5,478)	0.12/0.25 (99.8/98.6)	0.25/1 (97.8)	2/8 (96.6)	0.5/2 (97.4)	8/128 (79.4)	0.5/8 (81.2)
MDR (805)	0.12/0.5 (99.1/95.5)	1/4 (85.1)	8/16 (78.6)	2/8 (83.9)	128/>128 (11.1)	8/32 (18.0)
DTR (190)	0.12/1 (98.4/91.6)	2/8 (65.3)	8/32 (61.6)	4/>16 (70.5)	>128/>128 (0.0)	16/32 (0.0)
XDR (116)	0.12/2 (96.6/88.8)	4/>8 (24.1)	8/>32 (56.0)	4/>16 (59.5)	128/>128 (2.6)	16/>32 (0.0)
Carbapenem-nonsusceptible (1,238)	0.12/0.5 (99.4/96.8)	1/2 (90.1)	4/16 (87.1)	1/4 (90.1)	32/>128 (49.3)	8/32 (16.6)
Carbapenemase-positive[c] (13)	0.5/2 (100/84.6)	>8/>8 (23.1)	>32/>32 (23.1)	>16/>16 (7.7)	128/>128 (15.4)	>32/>32 (0.0)
Carbapenemase-negative (1,225)	0.12/0.5 (99.4/97.0)	1/2 (90.8)	4/16 (87.8)	1/4 (91.0)	32/>128 (49.7)	8/32 (16.8)

Abbreviations: FDC, cefiderocol; IMR, imipenem-relebactam; CZA, ceftazidime-avibactam; C/T, ceftolozane-tazobactam; P/T, piperacillin-tazobactam; MER, meropenem.
[a] MDR, multidrug resistance classified as non-susceptible (CLSI breakpoints) to ≥3 drug classes; DTR, difficult-to-treat resistance, defined as non-susceptible (CLSI) to piperacillin-tazobactam, ceftazidime, cefepime, aztreonam, meropenem, imipenem, ciprofloxacin, and levofloxacin; XDR, extensively drug resistant, defined as non-susceptible (CLSI) to all but 2 or fewer drug classes; carbapenem-nonsusceptible, isolates non-susceptible (MIC values ≥4 mg/L) to imipenem and/or meropenem based on CLSI criteria.
[b] Cefiderocol MIC results were interpreted according to the CLSI/FDA criteria, whereas comparator agent MIC values were interpreted based on CLSI criteria.
[c] Includes bla_{GES-5} (2), bla_{IMP-1} (1), bla_{IMP-13} (1), bla_{IMP-83} (1), bla_{KPC-2} (1), bla_{NDM-1} (2), bla_{VIM-1} (1), and bla_{VIM-2} (4).

Conclusions

- Cefiderocol showed potent activity against *P. aeruginosa* clinical isolates from US hospitals, including resistant subsets.
 - Resistant subsets included those with DTR and carbapenem-nonsusceptible phenotypes, against which cefiderocol showed activity higher than comparator agents.
 - Cefiderocol was also the most active against the small subset of isolates carrying carbapenemase genes.
- These data demonstrate cefiderocol *in vitro* activity against *P. aeruginosa* resistant subsets, for which antibiotic treatment options are limited.

Acknowledgments

This research and poster presentation were sponsored by Shionogi & Co., Ltd.

References

Clinical and Laboratory Standards Institute. 2018. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. *M07 11th Edition*. Wayne, PA, USA.

Clinical and Laboratory Standards Institute. 2024. Performance standards for antimicrobial susceptibility testing. *M100 34th Edition*. Wayne, PA, USA.

FDA Susceptibility Test Interpretive Criteria: https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed April 2024.

Karlowsky JA, Hackel MA, Takemura M, Yamano Y, Echols R, Sahm DF. 2022. *In vitro* susceptibility of Gram-negative pathogens to cefiderocol in five consecutive annual multinational SIDERO-WT Surveillance Studies, 2014 to 2019. *Antimicrob Agents Chemother*. 66: e0199021.

Mendes RE, Jones RN, Woosley LN, Cattoir V, Castanheira M. 2019. Application of next-generation sequencing for characterization of surveillance and clinical trial isolates: Analysis of the distribution of β-lactamase resistance genes and lineage background in the United States. *Open Forum Infect Dis* 6: S69–S78.

Ong'uti S, Czech M, Robilotti E, Holubar M. 2022. Cefiderocol: A new cephalosporin stratagem against multidrug resistant Gram-negative bacteria. *Clin Infect Dis*. 74: 1303–1312.

Contact

element

Rodrigo E. Mendes, Ph.D.
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo.mendes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_25-SHI-03_P2_PSA.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia