P-1606: Clearance of Infectious Virus in COVID-19 Patients Treated With Ensitrelvir Across Vaccination Status, Age, and Risk Subgroups: Exploratory Analyses From the SCORPIO-HR Study

Florin Draica[1], Anne Luetkemeyer[2], Kara W. Chew[3], Judith Currier[3], Davey Smith[4], Simon D. Portsmouth[5], Aeron C. Hurt[6], Takahiro Hasegawa[7], Safwan Kezbor[5], Daniel Verdi[1], Takeki Uehara[7]

[1]Medical Affairs, Shionogi Inc., Florham Park, NJ, USA; [2]Zuckerberg San Francisco General, University of California San Francisco, San Francisco, CA, USA; [3]David Geffen School of Medicine at University of California Los Angeles, Los Angeles, CA, USA; [4]Antiviral Research Center, University of California, San Diego, CA, USA; [5]Clinical Development, Shionogi Inc., Florham Park, NJ, USA; [6]Clinical Development, Shionogi & Co., Ltd., London, England, UK; [7]Drug Development and Regulatory Science Division, Shionogi & Co., Ltd., Osaka, Japan.

Shionogi Inc.
Address: 400 Campus Drive, Florham Park, NJ 07932, USA

Email ID: Florin.Draica@Shionogi.com
Mobile: +1-718-288-8349

Background

- While many antiviral studies for COVID-19 have relied on molecular methods to assess viral load, these methods may detect viral RNA even after the infectious virus has been cleared[1]
- In contrast, viral culture provides a direct measure of infectious viral shedding, potentially offering more accurate insights into transmission potential[1]
- Ensitrelvir is an oral SARS-CoV-2 3C-like (M^{pro}) protease inhibitor approved in Japan for the treatment of mild-to-moderate SARS-CoV-2 infection[2-5]
 - It has demonstrated consistent and potent antiviral efficacy, reducing both RNA copy number and infectious virus titer in those infected with SARS-CoV-2[6-8]
- In SCORPIO-HR, a phase 3 trial in nonhospitalized symptomatic adults with COVID-19, ensitrelvir treatment led to a reduction in mean RNA copy number and infectious virus titers in a higher proportion of participants by day 4 compared with placebo[8]

Objective

- This exploratory analysis of the SCORPIO-HR study assessed the effect of ensitrelvir vs placebo on the clearance of infectious virus on day 4 in participants with a day 1 positive viral culture across different subgroups

Methods

Study design[8]

- Multicenter, randomized, double-blind, placebo-controlled, phase 3 study (NCT05305547)
- Conducted in 16 countries in Africa, Asia, Europe, North America, and South America
- Study period: August 2022 to December 2023

Study design

SCORPIO-HR: Study design

A multicenter, randomized, double-blind, placebo-controlled study in 16 countries globally

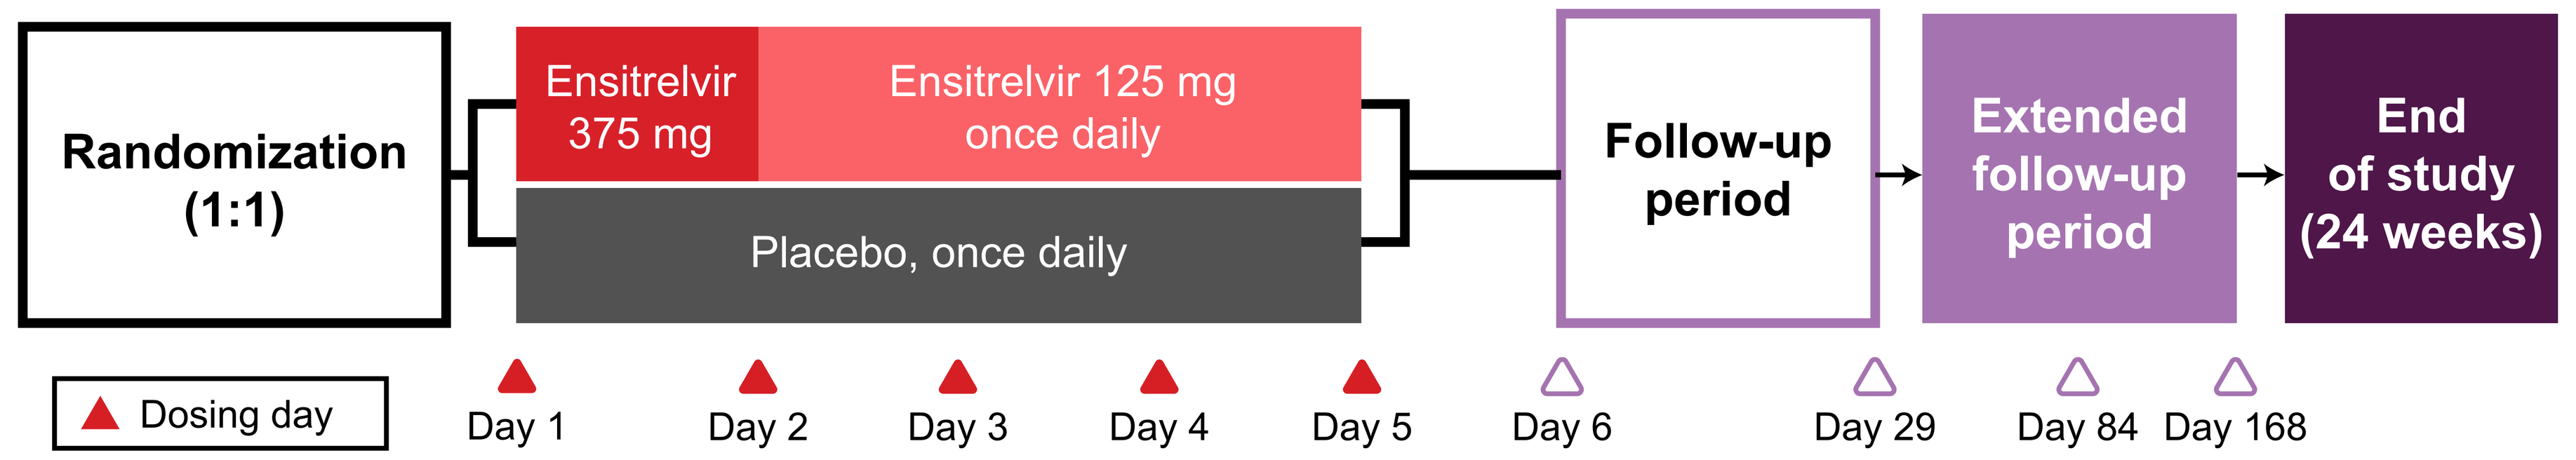

Randomization (1:1)
Ensitrelvir 375 mg
Ensitrelvir 125 mg once daily
Placebo, once daily
Follow-up period
Extended follow-up period
End of study (24 weeks)
Dosing day
Day 1
Day 2
Day 3
Day 4
Day 5
Day 6
Day 29
Day 84
Day 168

Key study eligibility criteria[8]

- Age ≥18 years
- Laboratory-confirmed SARS-CoV-2 infection
 - This post hoc analysis was limited to participants with detectable viral culture (>lower limit of quantification [LLoQ=1.0]) on day 1
- Symptom onset ≤3 days (initially ≤5 days) before randomization

Endpoint for exploratory analysis

- Proportion of participants with a day 1 positive viral culture who had a negative viral culture (<LLoQ) on day 4
 - Subgroup results were reported by risk status for severe COVID-19 (high and standard risk), age (<65 and ≥65 years), and COVID-19 vaccination status (vaccinated and unvaccinated)
- As this was an exploratory post hoc analysis, no formal statistical testing was performed

Viral culture procedure

- Quantitative viral culture (LLoQ=1.0) was conducted centrally at Cerba Research (formerly Viroclinics-DDL, Rotterdam, Netherlands) using nasopharyngeal swabs collected on days 1, 4, 8, and 16
- Samples were stabilized (transferred to Universal Transport Medium, aliquoted, and frozen) at designated local facilities within 48 hours of collection and shipped to the central laboratory for viral culture
- Viral titer was measured by virus-induced cytopathic effects in VeroE6/TMPRSS2 cells

Results

- In SCORPIO-HR, 28.7% (601/2093) of participants had positive viral cultures at enrollment
- Among the 567 participants with viral culture data available at day 4, 95.5% (274/287) in the ensitrelvir group vs 75.0% (210/280) in the placebo group were viral culture negative
- Baseline characteristics were mostly similar between the two groups (**Table 1**)

Table 1. Characteristics of participants with day 1 positive viral culture and available day 4 viral culture data

	Ensitrelvir* (N=287)	Placebo* (N=280)
Age, n (%)		
<65 years	267 (93.0)	257 (91.8)
≥65 years	20 (7.0)	23 (8.2)
Sex, n (%)		
Male	104 (36.2)	114 (40.7)
Female	183 (63.8)	166 (59.3)
Risk status†, n (%)		
High risk	88 (30.7)	99 (35.4)
Standard risk	199 (69.3)	181 (64.6)
COVID-19 vaccination‡, n (%)		
Yes	218 (76.5)§	229 (81.8)
No	67 (23.5)§	51 (18.2)

*Among the 601 participants enrolled with positive viral cultures on day 1 (ensitrelvir: n=309; placebo: n=292), 34 participants without viral culture data on day 4 were excluded from this summary. These participants were excluded from the analysis assuming that they were completely missing at random.[8] †High-risk participants were defined as individuals aged ≥65 years or aged ≥18 years with ≥1 of the following: obesity (BMI ≥30 kg/m²), diabetes mellitus, hypertension or cardiovascular/lung/kidney disease requiring daily therapy, congenital heart disease, Down syndrome, sickle cell disease, or immunocompromising conditions/immunosuppressive treatments (e.g., cancer therapy, hematologic malignancy, transplant history, HIV with CD4+ <200 cells/mm³, primary immunodeficiency, immunosuppressive medications). Standard-risk participants were defined as those aged 18–64 years with none of these risk factors. ‡Participants were classified as unvaccinated if they responded "No" to the question "Has the subject taken COVID-19 vaccines?" or if they had received vaccination but had not completed the primary series of COVID-19 vaccinations. §Vaccination status was missing in two enistrelvir-treated participants.

- Across all subgroups, a greater proportion of participants in the ensitrelvir vs placebo group achieved a negative viral culture on day 4 (**Figure 1**)

Figure 1. Proportion of participants with negative SARS-CoV-2 viral culture on day 4

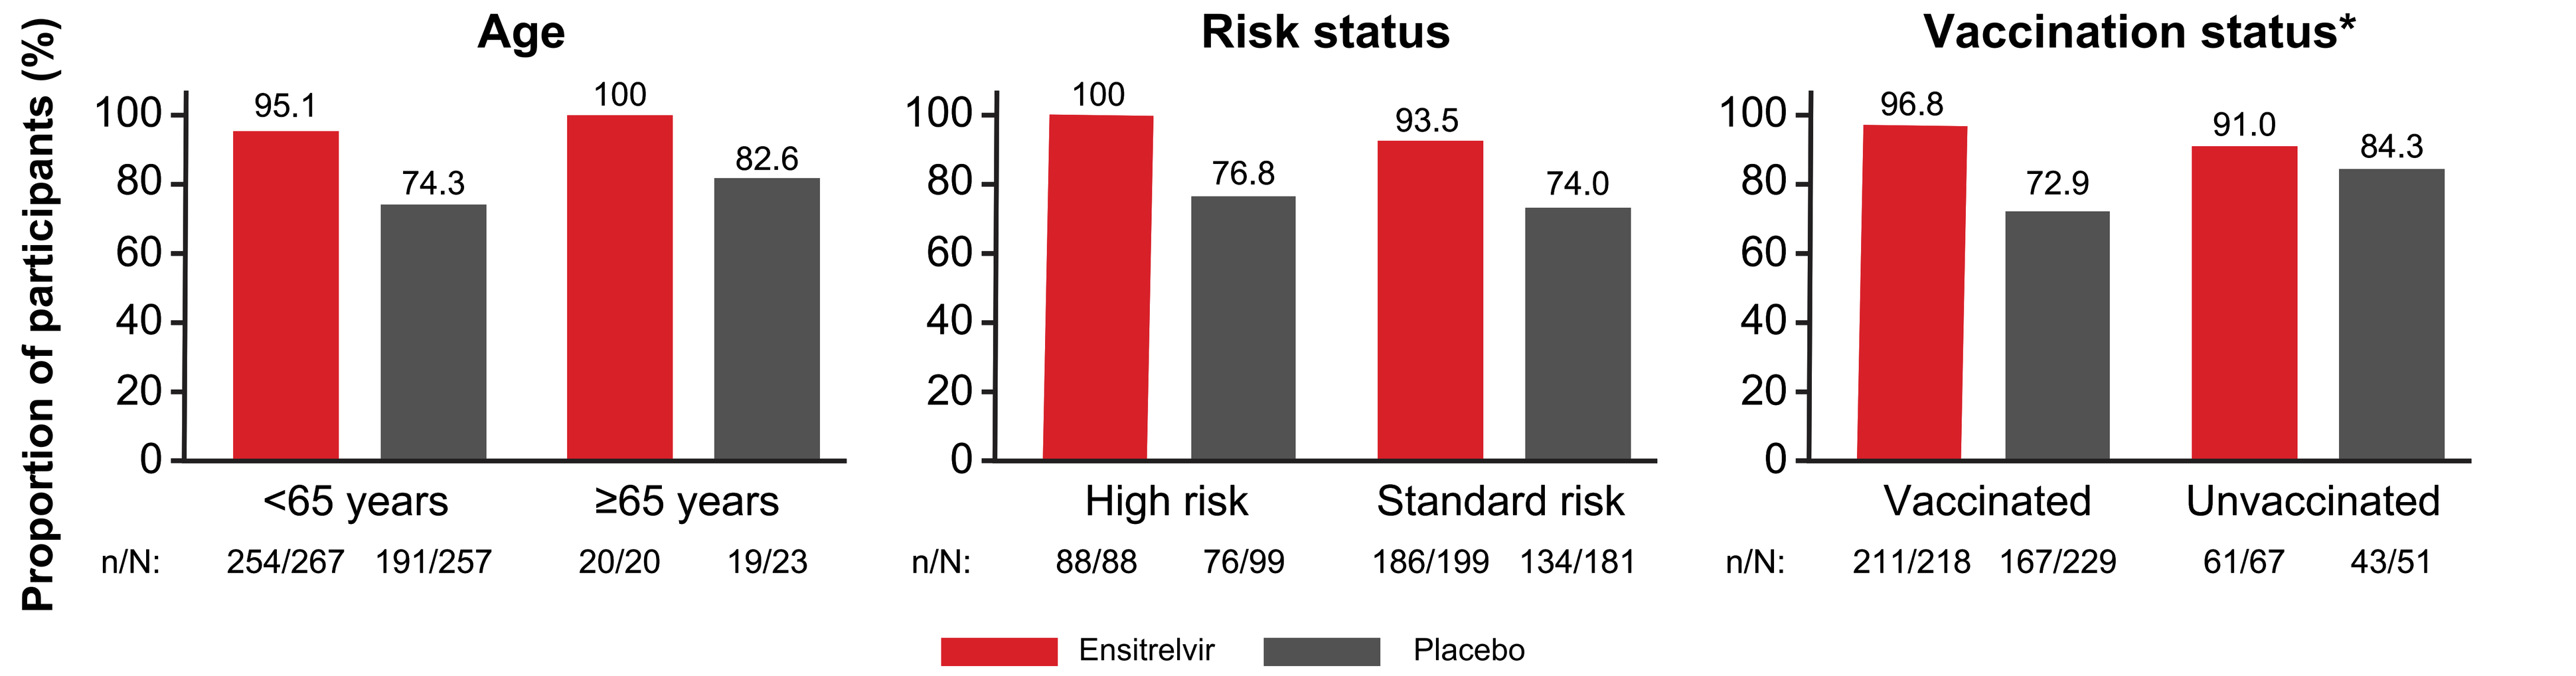

Proportion of participants (%)
Age
Risk status
Vaccination status*
100
80
60
40
20
0
95.1
74.3
100
82.6
<65 years
≥65 years
n/N:
254/267
191/257
20/20
19/23
100
76.8
93.5
74.0
High risk
Standard risk
88/88
76/99
186/199
134/181
96.8
72.9
91.0
84.3
Vaccinated
Unvaccinated
211/218
167/229
61/67
43/51
Ensitrelvir
Placebo

*Participants were classified as unvaccinated if they responded "No" to the question "Has the subject taken COVID-19 vaccines?" or if they had received vaccination but had not completed the primary series of COVID-19 vaccinations. n=number of participants with viral clearance on day 4; N=total number of participants in the group.

- Among vaccinated participants, clearance of infectious virus was consistently higher with ensitrelvir than with placebo, regardless of risk status and age (**Figure 2**)

Figure 2. Proportion of participants with negative SARS-CoV-2 viral culture on day 4 by vaccination status in combination with age and risk status

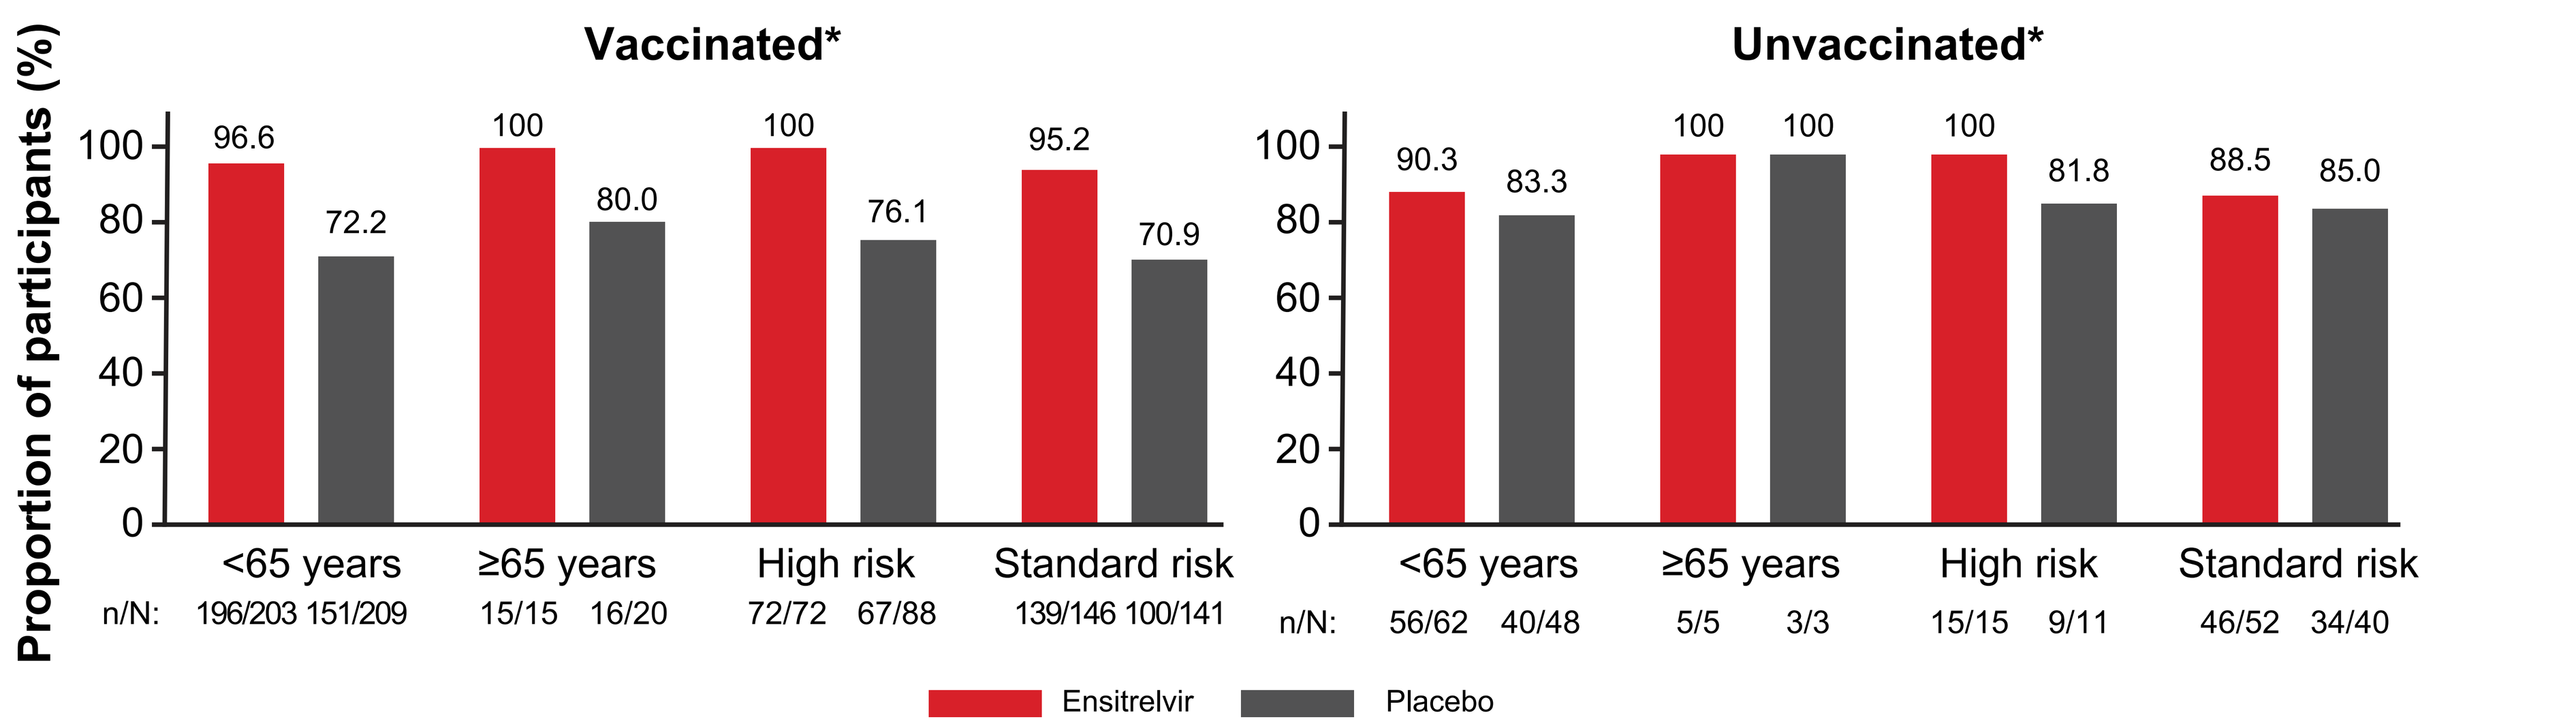

Proportion of participants (%)
Vaccinated*
Unvaccinated*
100
80
60
40
20
0
96.6
72.2
100
80.0
100
76.1
95.2
70.9
<65 years
≥65 years
High risk
Standard risk
n/N:
196/203 151/209
15/15 16/20
72/72 67/88
139/146 100/141
90.3
83.3
100
100
100
81.8
88.5
85.0
56/62 40/48
5/5 3/3
15/15 9/11
46/52 34/40
Ensitrelvir
Placebo

*Participants were classified as unvaccinated if they responded "No" to the question "Has the subject taken COVID-19 vaccines?" or if they had received vaccination but had not completed the primary series of COVID-19 vaccinations. n=number of participants with viral clearance on day 4; N=total number of participants in the group.

Figure 3. Forest plot of risk ratios for SARS-CoV-2 viral culture negativity on day 4

Subgroups	Ensitrelvir, % (n/N)	Placebo, % (n/N)	Risk ratio (95% CI)*
<65 yrs	95.1 (254/267)	74.3 (191/257)	1.28 (1.06, 1.55)
≥65 yrs	100 (20/20)	82.6 (19/23)	1.21 (0.61, 2.40)
Male	93.3 (97/104)	70.2 (80/114)	1.33 (0.98, 1.81)
Female	96.7 (177/183)	78.3 (130/166)	1.24 (0.98, 1.56)
HR	100 (88/88)	76.8 (76/99)	1.30 (0.95, 1.79)
SR	93.5 (186/199)	74.0 (134/181)	1.26 (1.01, 1.59)
Vaccinated	96.8 (211/218)	72.9 (167/229)	1.33 (1.08, 1.64)
Unvaccinated	91.0 (61/67)	84.3 (43/51)	1.08 (0.72, 1.63)
Vaccinated <65 yrs	96.6 (196/203)	72.2 (151/209)	1.34 (1.08, 1.66)
Vaccinated ≥65 yrs	100 (15/15)	80.0 (16/20)	1.25 (0.58, 2.70)
Vaccinated HR	100 (72/72)	76.1 (67/88)	1.31 (0.93, 1.86)
Vaccinated SR	95.2 (139/146)	70.9 (100/141)	1.34 (1.03, 1.75)
Unvaccinated <65 yrs	90.3 (56/62)	83.3 (40/48)	1.08 (0.71, 1.67)
Unvaccinated ≥65 yrs	100 (5/5)	100 (3/3)	1.00 (0.19, 6.44)
Unvaccinated HR	100 (15/15)	81.8 (9/11)	1.22 (0.50, 3.17)
Unvaccinated SR	88.5 (46/52)	85.0 (34/40)	1.04 (0.65, 1.67)

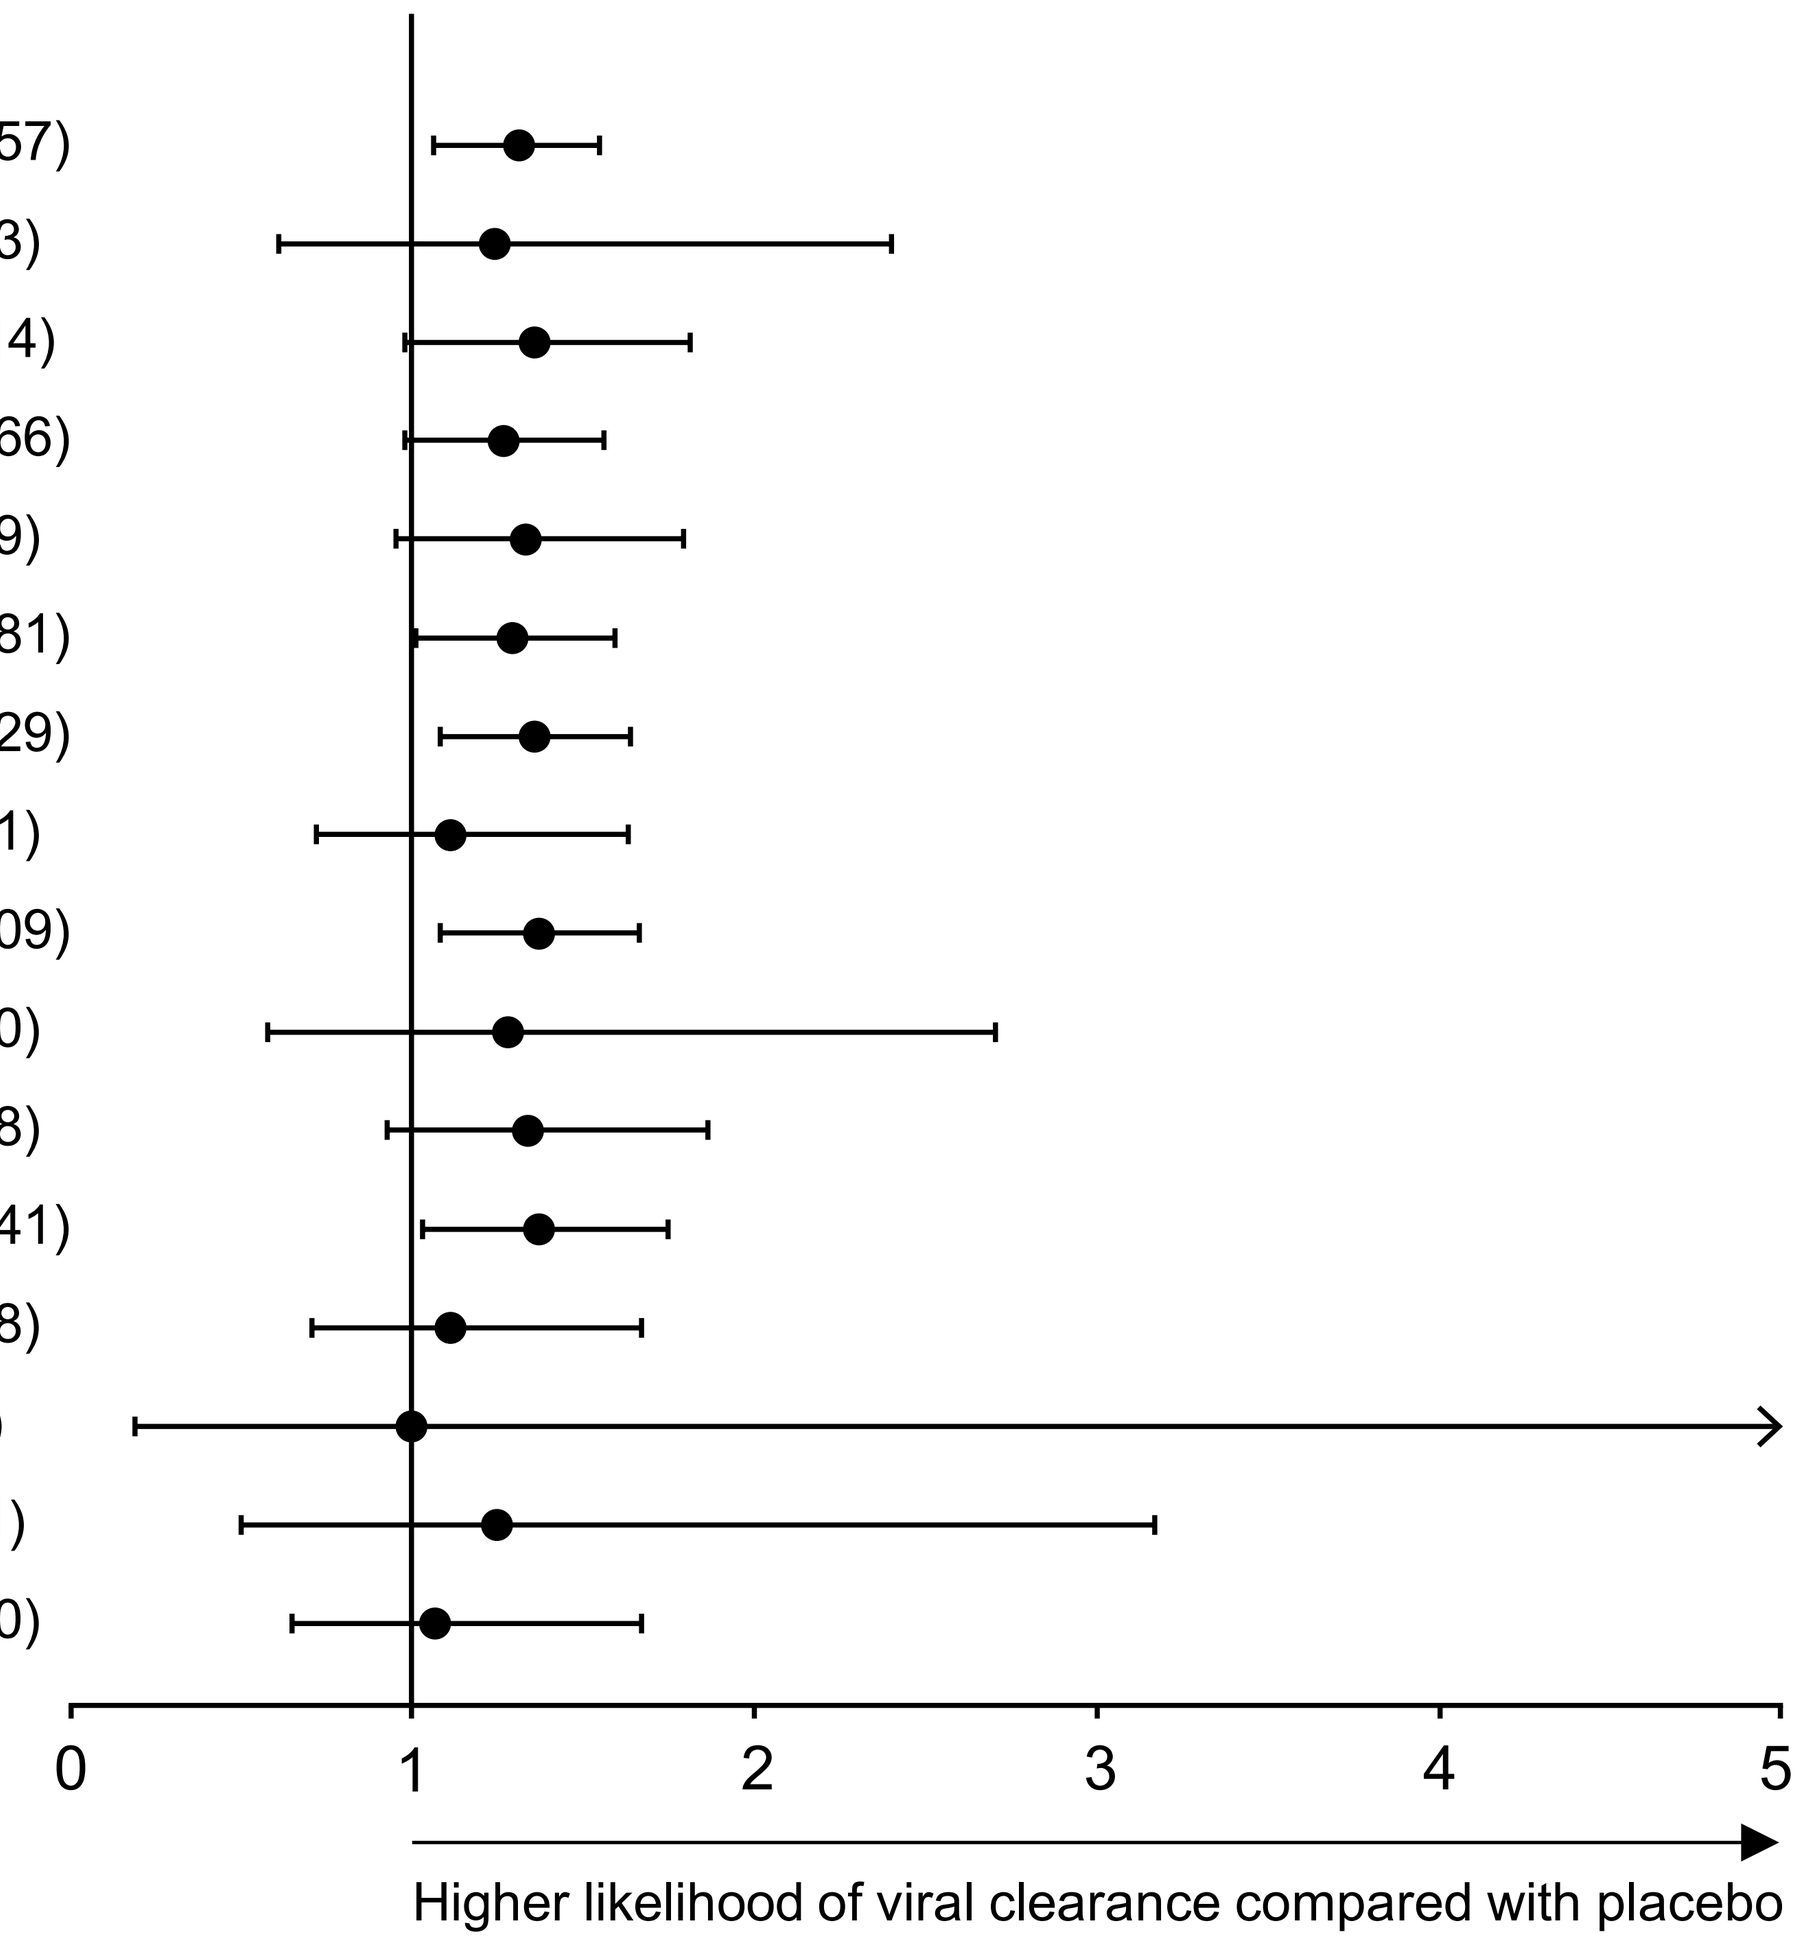

0
1
2
3
4
5
Higher likelihood of viral clearance compared with placebo

*The 95% CI of the risk ratio was obtained using the exact method.
CI, confidence interval; HR, high risk; SR, standard risk; yrs, years.

- By day 4, the following groups showed a numerically higher likelihood of achieving a negative viral culture when treated with ensitrelvir compared with placebo **(Figure 3)**: <65 years, standard risk, vaccinated, vaccinated <65 years, and vaccinated standard risk

Conclusion

- Ensitrelvir demonstrated consistent antiviral efficacy, as measured by clearance of infectious virus, across participant characteristics of vaccination status, age, and risk for severe disease

References

1. Puhach O, et al. *Nat Rev Microbiol*. 2023;21(3):147-161.
2. Kawashima S, et al. *Biochem Biophys Res Commun*. 2023;645:132-136.
3. Kuroda T, et al. *J Antimicrob Chemother.* 2023;78(4):946-952.
4. Nobori H, et al. *Antiviral Res*. 2024;224:105852.
5. Shionogi. https://www.shionogi.com/global/en/news/2024/03/20240305.html. Accessed August 19, 2025.
6. Mukae H, et al. *Antimicrob Agents Chemother*. 2022;66(10):e0069722.
7. Yotsuyanagi H, et al. *JAMA Netw Open.* 2024;7(2):e2354991.
8. Luetkemeyer AF, et al. *Clin Infect Dis.* 2025;80(6):1235-1244.

Abbreviations

BMI, body mass index; CD4, cluster of differentiation 4; CI, confidence interval; COVID-19, coronavirus disease 2019; HIV, human immunodeficiency virus; HR, high risk; RNA, ribonucleic acid; SARS-CoV-2, severe acute respiratory syndrome coronavirus 2; SR, standard risk; yrs, years

Acknowledgements

The authors thank all the participants involved in this study. This work was supported by the National Institute of Allergy and Infectious Diseases of the National Institute of Health (NIH) under Award numbers UM1AI068636, UM1AI068634, and UM1AI106701, and Shionogi & Co., Ltd.; SCORPIO-HR ClinicalTrials.gov number: NCT05305547. Medical writing support was provided by Gauri Pradhan, PhD, CMPP™, of Cactus Life Sciences (part of Cactus Communications) and funded by Shionogi Inc.

Disclosures

FD, SDP, SK, and **DV** are employees of Shionogi Inc (none own stock). **AL** received research grant support to the institution from NIH (for this study) and has received unrelated research grant support to the institution from Cepheid, Gilead, GSK, and Merck. **KWC** served as a consultant for Pardes Bioscience (concluded). **JC** received honoraria for service on an advisory board for Merck and Co. **DS** served as an advisor/consultant for Model Medicines, Obsidian, Bayer, Pharma Holdings, and Hyundai; served on the data safety monitoring board or advisory board for Gilead and Red Queen Therapeutics; was involved in the project startup of A2 Bio; has stock or stock options in Fluxergy, Linear Therapies, and Model Medicines; received grant/research support from the NIH; and received payment or honoraria for lectures from IAS USA. **ACH** is an employee of Shionogi BV (does not own stock). **TH** and **TU** are employees of Shionogi & Co., Ltd.

IDWeek 2025, October 19–22, 2025, Atlanta, GA