P-2044: Analysis of Post COVID-19 Condition After Ensitrelvir Treatment in Asymptomatic or Mild-Symptoms Only Patients in the SCORPIO Phase 2b/3 Study

Takumi Imamura[1], Hiroshi Yotsuyanagi[2], Norio Ohmagari[3], Yohei Doi[4], Masaya Yamato[5], Hiroki Sakaguchi[1], Hideki Yamanaka[1], Ryosuke Imaoka[1], Akimasa Fukushi[1], Genki Ichihashi[1], Yuko Tsuge[1], Takeki Uehara[1], Hiroshi Mukae[6]

[1]Drug Development and Regulatory Science Division, Shionogi & Co., Ltd., Osaka, Japan; [2]The Institute of Medical Science, The University of Tokyo, Tokyo, Japan; [3]Disease Control and Prevention Center, National Center for Global Health and Medicine, Tokyo, Japan; [4]Departments of Microbiology and Infectious Diseases, Fujita Health University School of Medicine, Toyoake, Japan; [5]Department of General Medicine and Infectious Diseases, Rinku General Medical Center, Izumisano, Japan; [6]Department of Respiratory Medicine, Nagasaki University Graduate School of Biomedical Sciences, Nagasaki, Japan

Shionogi & Co., Ltd.
Address: 8F, Nissay Yodoyabashi East, 3-3-13 Imabashi, Chuo-ku, Osaka 541-0042, Japan
E-mail: takumi.imamura@shionogi.co.jp
Phone no.: +81-6-6209-6907

INTRODUCTION

- PCC (Long COVID) is a multifaceted health concern observed in individuals with a history of SARS-CoV-2 infection [1]
 - PCC generally occurs late after the onset of COVID-19 and poses a negative impact on patients' daily lives [1]
 - PCC may manifest in patients with mild COVID-19 and may persist for more than 2 years [2]
- A correlation between SARS-CoV-2 viral persistence and the risk of PCC has been suggested [3]
 - Research is warranted to establish the efficacy of anti–SARS-CoV-2 antivirals in preventing PCC
- Ensitrelvir is an oral SARS-CoV-2 3C-like protease inhibitor approved in Japan and Singapore for the treatment of mild-to-moderate COVID-19 [4]
 - An exploratory analysis of the Phase 3 SCORPIO-SR study reported potential efficacy of ensitrelvir in reducing the risk of PCC among patients with mild-to-moderate COVID-19 [5]
- We assessed the efficacy of ensitrelvir in preventing PCC in patients with asymptomatic SARS-CoV-2 infection or mild COVID-19 symptoms only

METHODS

- This Phase 2b/3 trial was conducted as part of a seamless, multicenter, randomized, double-blind, placebo-controlled, Phase 2/3 study (Japan Registry of Clinical Trials identifier: jRCT2031210350) [6]
- This analysis included patients with asymptomatic SARS-CoV-2 infection or mild COVID-19 symptoms who were screened but not enrolled in the Phase 3 SCORPIO-SR study [5]
- PCC was defined based on the self-assessed general health level and severity of a prespecified set of symptoms
- Investigators and patients remained blinded on treatment assignment until Day 337

Figure 1. Study design

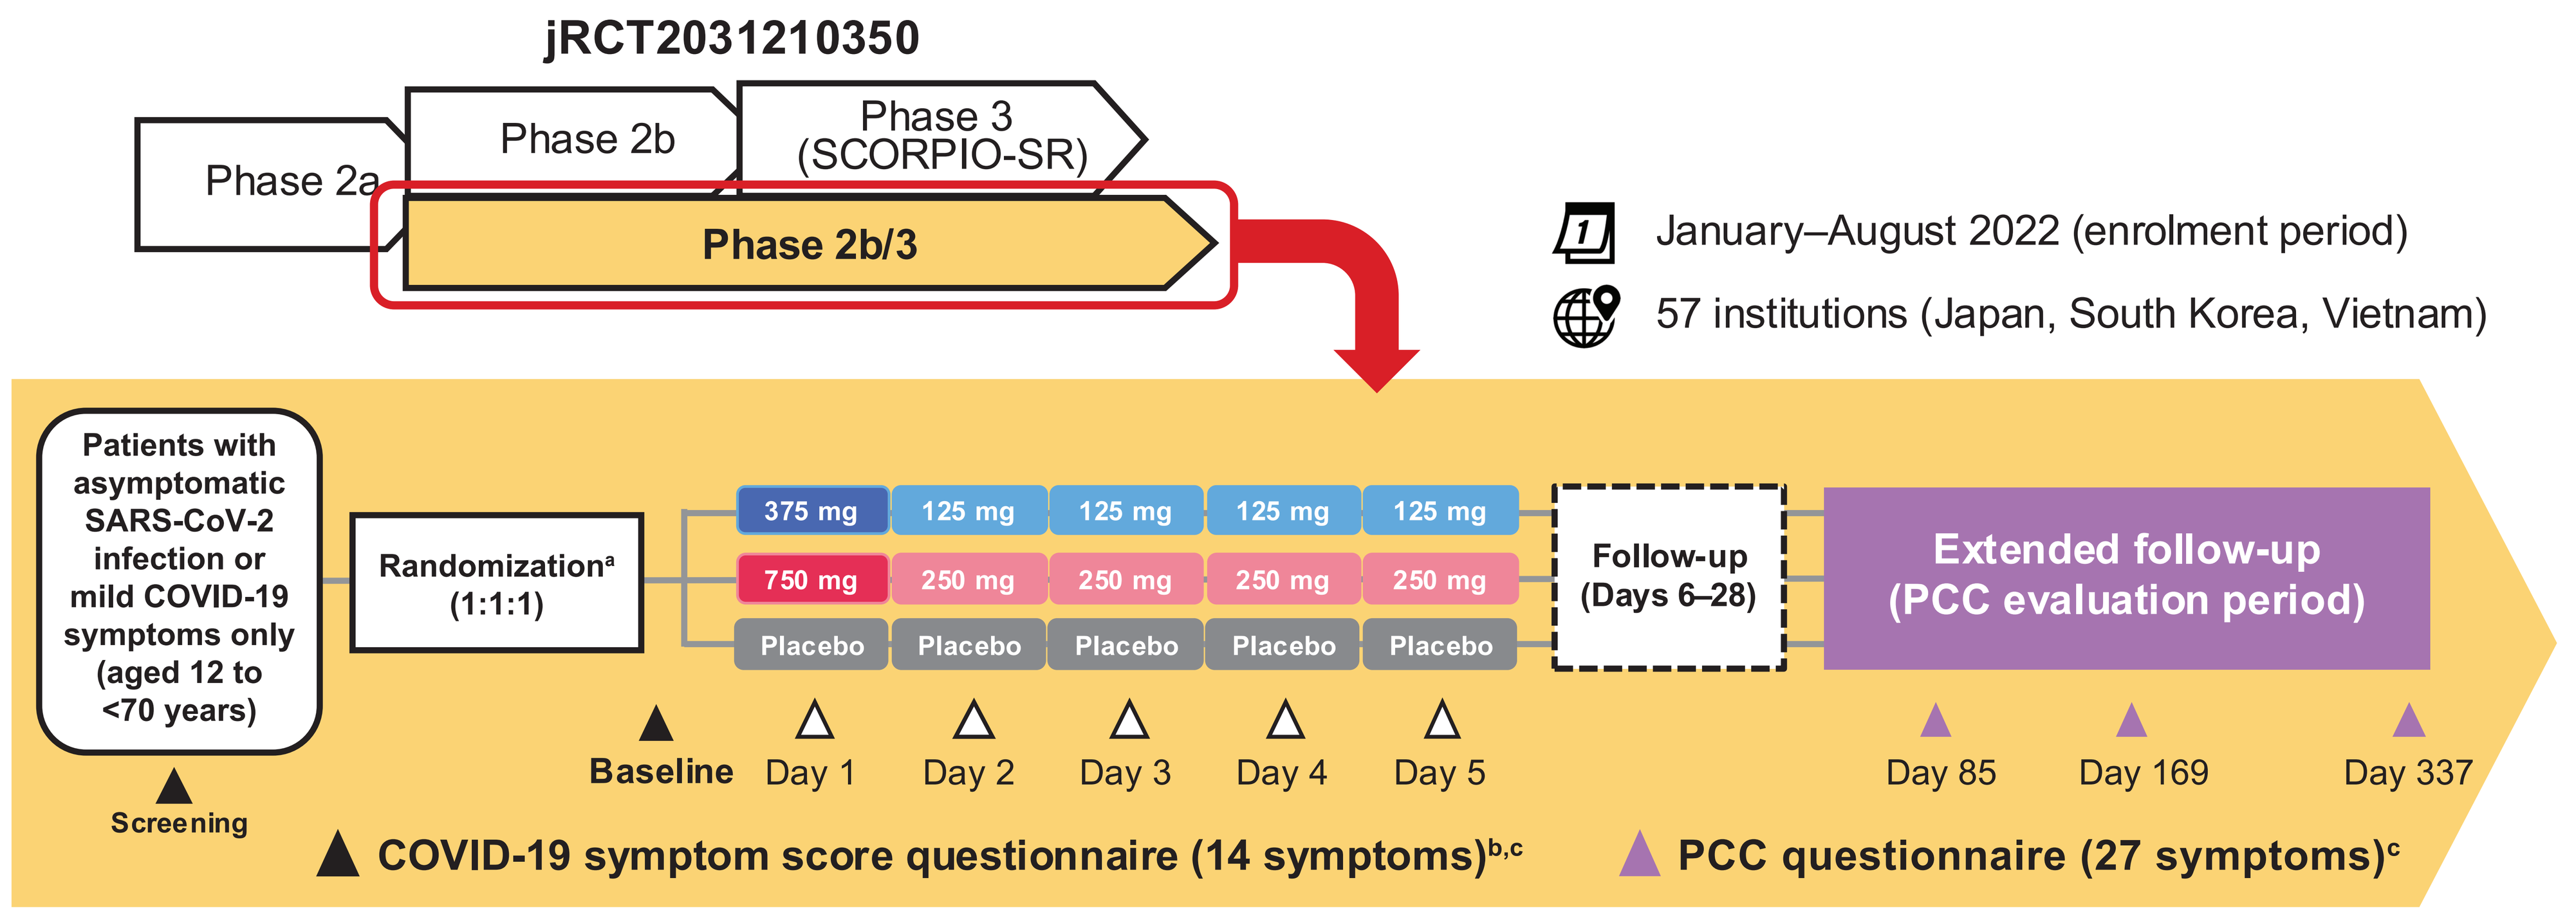
jRCT2031210350
Phase 2a
Phase 2b
Phase 3 (SCORPIO-SR)
Phase 2b/3
January–August 2022 (enrolment period)
57 institutions (Japan, South Korea, Vietnam)
Patients with asymptomatic SARS-CoV-2 infection or mild COVID-19 symptoms only (aged 12 to <70 years)
Screening
Randomization[a] (1:1:1)
375 mg
125 mg
750 mg
250 mg
Placebo
Follow-up (Days 6–28)
Extended follow-up (PCC evaluation period)
Baseline
Day 1
Day 2
Day 3
Day 4
Day 5
Day 85
Day 169
Day 337
COVID-19 symptom score questionnaire (14 symptoms)[b,c]
PCC questionnaire (27 symptoms)[c]

[a]≤120 hours from the onset of COVID-19 symptoms.
[b]The questionnaire was used for screening and administered at baseline and throughout the study period until Day 21. Only data for baseline are presented in this analysis.
[c]Patients self-assessed the symptoms and recorded the results in an electronic diary.

PCC questionnaire (excerpt)

- Please choose the response that best describes the severity of your COVID-19 symptoms over the past 4 weeks (No symptoms, Mild, Moderate, or Severe)
- Please choose the response that best describes your general physical health over the past 4 weeks (Excellent, Very good, Good, Fair, or Poor)
- Have you returned to your usual (pre-COVID) health? (Yes or No)
- Please rate the severity of each of the 27 symptoms over the past 4 weeks (Absent, Mild, Moderate, or Severe)

Definitions of PCC

- General health condition self-reported as "not returned to pre-COVID health level" AND
- ≥1 mild, moderate, or severe symptom among the symptoms listed below:
 - 27 symptoms: Stuffy nose, runny nose, sore throat, shortness of breath, cough, low energy or tiredness, muscle or body aches, headache, chills or shivering, feeling hot or feverish, nausea, vomiting, diarrhea, loss of smell, loss of taste, muscle weakness, hair loss, palpitations, joint pain, decreased appetite, dizziness, chest pain, skin rash, difficulty with concentration and thinking, difficulty in reasoning and solving problems, memory loss, or insomnia
 - 4 neurological symptoms: Difficulty with concentration and thinking, difficulty in reasoning and solving problems, memory loss, or insomnia
 - 14 symptoms: Stuffy or runny nose, sore throat, shortness of breath, cough, low energy or tiredness, muscle or body aches, headache, chills or shivering, feeling hot or feverish, nausea, vomiting, diarrhea, loss of smell, or loss of taste
 - 5 WHO symptoms (fatigue, shortness of breath, difficulty with concentration and thinking, difficulty in reasoning and solving problems, or memory loss [1]), loss of smell, or loss of taste

RESULTS

Table 1. Baseline characteristics

- In total, 384 patients were included in the analysis (mild COVID-19 symptoms only, n=351; asymptomatic SARS-CoV-2 infection, n=33)
- Patient characteristics were generally balanced across treatment groups
- Approximately 90% of the patients had SARS-CoV-2 vaccination history

		Ensitrelvir 125 mg N=124	Ensitrelvir 250 mg N=129	Placebo N=131
Sex	Male	70 (56.5%)	79 (61.2%)	67 (51.1%)
Age, years	Mean (SD)	38.8 (11.9)	41.6 (13.4)	39.1 (13.3)
BMI	Mean (SD)	22.80 (3.25)	23.31 (3.48)	22.96 (4.16)
Smoking habit	Yes	22 (17.7%)	23 (17.8%)	23 (17.6%)
Randomization in <72 hours of onset	Yes	47 (37.9%)	52 (40.3%)	57 (43.5%)
≥1 SARS-CoV-2 vaccine dose	Yes	115 (92.7%)	116 (89.9%)	122 (93.1%)
Total score of 14 symptoms at baseline[a]	Mean (SD)	4.1 (2.6)	4.2 (3.4)	3.9 (2.7)

Patients in the ITT population who responded to the PCC questionnaire at any of the three time points (Day 85, 169, or 337) are included.
[a]Patients self-rated their 14 symptoms on a 4-point (None=0, Mild=1, Moderate=2, or Severe=3) or 3-point (sense of smell and sense of taste only; Same as usual=0, Less than usual=1, or No sense of smell or taste=2) scale.
The scores for each symptom were added to calculate the total score for the 14 COVID-19 symptoms.

Table 2. Self-reported COVID-19 severity and physical health status

- On Day 337, 4.3%, 3.4%, and 8.8% of patients in the ensitrelvir 125 mg, ensitrelvir 250 mg, and placebo groups, respectively, reported that their health status had not returned to usual levels

Time Point	Endpoint	Ensitrelvir 125 mg N=124	Ensitrelvir 250 mg N=129	Placebo N=131
Day 85	Mild, moderate, or severe COVID-19 symptoms over the past 4 weeks	5.3% (6/114)	1.7% (2/120)	9.8% (12/122)
	Poor general physical health over the past 4 weeks	1.8% (2/114)	0.8% (1/120)	3.3% (4/122)
	Health status not returning to usual levels	7.0% (8/114)	3.3% (4/120)	13.1% (16/122)
Day 169	Mild, moderate, or severe COVID-19 symptoms over the past 4 weeks	4.3% (5/117)	4.8% (6/125)	6.2% (8/129)
	Poor general physical health over the past 4 weeks	0.9% (1/117)	1.6% (2/125)	3.1% (4/129)
	Health status not returning to usual levels	5.1% (6/117)	2.4% (3/125)	7.8% (10/129)
Day 337	Mild, moderate, or severe COVID-19 symptoms over the past 4 weeks	0.9% (1/115)	2.6% (3/117)	3.2% (4/125)
	or general physical health over the past 4 weeks	1.7% (2/115)	2.6% (3/117)	4.8% (6/125)
	Health status not returning to usual levels	4.3% (5/115)	3.4% (4/117)	8.8% (11/125)

Patients in the ITT population who responded to the PCC questionnaire at any of the three time points (Day 85, 169, or 337) are included.

Figure 2. Proportion of patients with PCC and risk reduction versus placebo

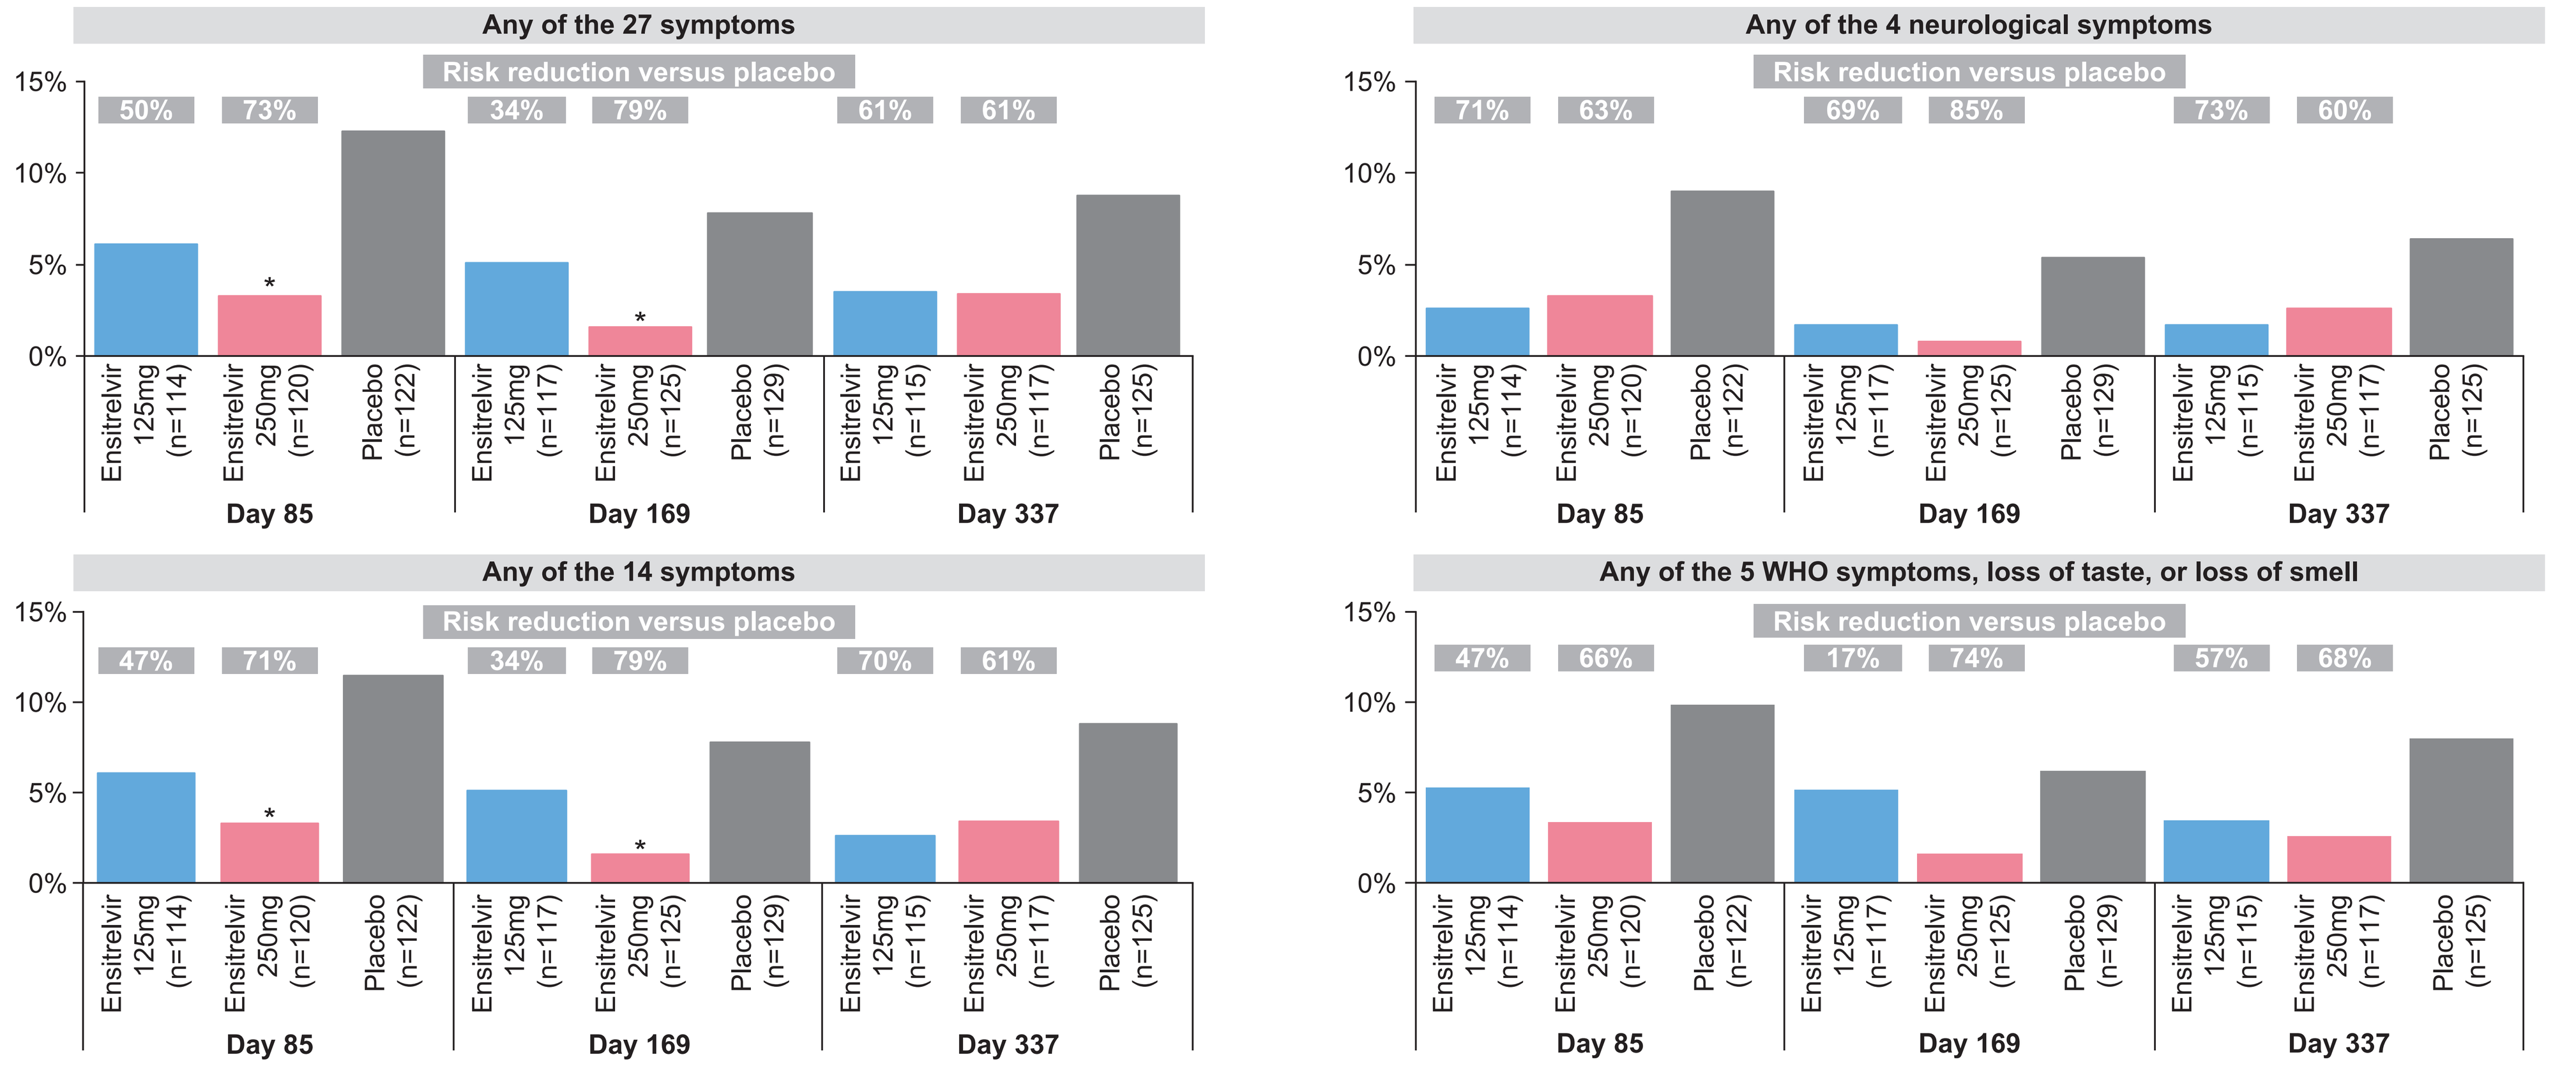
Any of the 27 symptoms
Risk reduction versus placebo
50% 73% 34% 79% 61% 61%
Any of the 4 neurological symptoms
Risk reduction versus placebo
71% 63% 69% 85% 73% 60%
Any of the 14 symptoms
Risk reduction versus placebo
47% 71% 34% 79% 70% 61%
Any of the 5 WHO symptoms, loss of taste, or loss of smell
Risk reduction versus placebo
47% 66% 17% 74% 57% 68%
15%
10%
5%
0%
Ensitrelvir 125mg (n=114)
Ensitrelvir 250mg (n=120)
Placebo (n=122)
Ensitrelvir 125mg (n=117)
Ensitrelvir 250mg (n=125)
Placebo (n=129)
Ensitrelvir 125mg (n=115)
Ensitrelvir 250mg (n=117)
Placebo (n=125)
Day 85
Day 169
Day 337

Data are summarized for patients who completed the PCC questionnaire for at least one timepoint in the ITT population. *p<0.05 (Fisher's exact test)

- Among the 33 asymptomatic patients (ensitrelvir 125 mg, n=9; ensitrelvir 250 mg, n=12; placebo, n=12), 1 participant in the ensitrelvir 125 mg group experienced PCC (any of the 27 symptoms) on Day 169 and Day 337. In the placebo group, 1 participant experienced PCC (any of the 27 symptoms) on Day 85 and Day 169
- On Day 337, ensitrelvir 125 mg and 250 mg groups showed a trend towards risk reduction versus placebo for PCC (statistically non-significant)

Figure 3. Relative risk reduction versus placebo for the most common individual symptoms of PCC

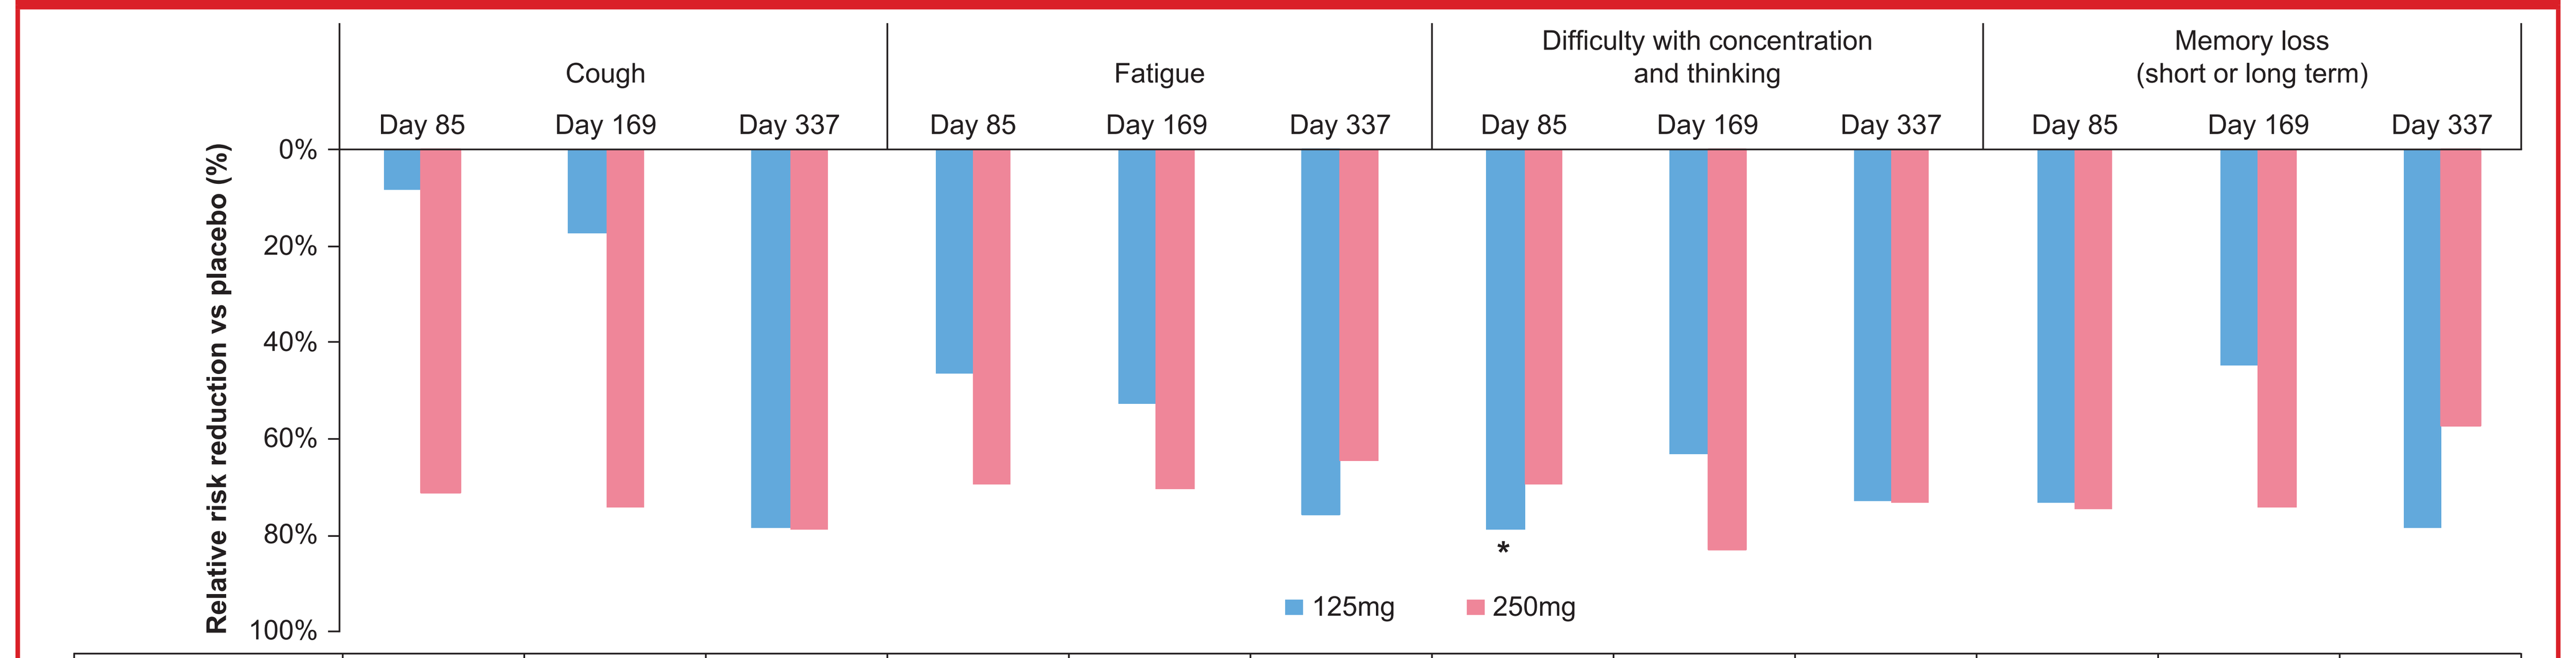
Cough
Fatigue
Difficulty with concentration and thinking
Memory loss (short or long term)
Day 85
Day 169
Day 337
Relative risk reduction vs placebo (%)
0%
20%
40%
60%
80%
100%
125mg
250mg

	Cough Day 85	Cough Day 169	Cough Day 337	Fatigue Day 85	Fatigue Day 169	Fatigue Day 337	Difficulty with concentration and thinking Day 85	Difficulty with concentration and thinking Day 169	Difficulty with concentration and thinking Day 337	Memory loss Day 85	Memory loss Day 169	Memory loss Day 337
Ensitrelvir 125 mg	6/114	3/117	1/115	5/114	3/117	2/115	2/114	2/117	2/115	2/114	2/117	1/115
Ensitrelvir 250 mg	2/120	1/125	1/117	3/120	2/125	3/117	3/120	1/125	2/117	2/120	1/125	2/117
Placebo	7/122	4/129	5/125	10/122	7/129	9/125	10/122	6/129	8/125	8/122	4/129	5/125

Data are summarized for patients who completed the PCC questionnaire for at least one timepoint in the ITT population.
PCC symptoms reported in at least one time point with an incidence ≥5% in the placebo group are presented. *p<0.05 (Fisher's exact test)

- Ensitrelvir 125 mg and 250 mg groups also showed a trend towards risk reduction versus placebo for the most common individual symptoms of PCC

CONCLUSIONS

- PCC was observed even in patients with asymptomatic SARS-CoV-2 infection and those with mild COVID-19 symptoms only. It was also suggested that ensitrelvir treatment may reduce PCC in such patients with asymptomatic/mild disease

ABBREVIATIONS

BMI, body mass index; **COVID-19**, coronavirus disease 2019; **ITT,** intention-to-treat; **PCC,** post COVID-19 condition; **SARS-CoV-2,** severe acute respiratory syndrome coronavirus 2; **SD,** standard deviation; **WHO,** World Health Organization

REFERENCES

[1] Soriano JB, et al. *Lancet Infect Dis.* 2022;22:e102–7. [2] Morioka S, et al. *J Infect Chemother.* 2024;30(8):734–40. [3] Yang C, et al. *Lancet Respir Med.* 2023;11(6):504–6. [4] Syed YY. *Drugs.* 2024;84(6):721–8. [5] Yotsuyanagi H, et al. *Antiviral Res.* 2024:229;105958. [6] Ohmagari N, et al. *Influenza Other Respir Viruses.* 2024;18(6):e13338.

DISCLOSURES

T. Imamura, H. Sakaguchi, H, Yamanaka, R. Imaoka, A. Fukushi, G. Ichihashi, Y. Tsuge, and T. Uehara are full-time employees of Shionogi & Co., Ltd. H. Yotsuyanagi, Y. Doi, M. Yamato, and H. Mukae are advisory board members and have received funding from Shionogi & Co., Ltd. N. Ohmagari is an advisory board member of Shionogi & Co., Ltd. without compensation.

ACKNOWLEDGMENTS

This Phase 2b/3 study (jRCT2031210350) was sponsored by Shionogi & Co., Ltd. and financially supported by the Organization of the Ministry of Health, Labour and Welfare of Japan. The authors and research team thank all patients involved in this study and Tomoko Tsushima and Tsukasa Horiyama (Shionogi & Co., Ltd.) for preparing technical-support documents. Medical writing support was provided by Mami Hirano, MSc, ISMPP CMPP, of Cactus Life Sciences (part of Cactus Communications) and funded by Shionogi & Co., Ltd.

IDWeek 2024, October 16–19, 2024, Los Angeles, CA