IDWEEK 2024 | Poster #P1360

Activity of Cefiderocol against Carbapenem Non-susceptible Enterobacterales, Including Molecularly Characterized Multidrug-Resistant Clinical Isolates, Causing Infections in United States Hospitals (2020–2023)

RE Mendes, D Beekman, JM Maher, JH Kimbrough, HS Sader, M Castanheira
Element Iowa City (JMI Laboratories), North Liberty, IA, USA

Introduction

- β-lactamase genes, such as those encoding class A and D serine carbapenemases and class B metallo-β-lactamases (MBL), contribute to the emergence and dissemination of carbapenem nonsusceptible Enterobacterales.
- Cefiderocol is approved by the US Food and Drug Administration (FDA) for the treatment of complicated urinary tract infections, including pyelonephritis, as well as hospital-acquired bacterial pneumonia and ventilator-associated bacterial pneumonia.
- Cefiderocol is a siderophore cephalosporin with broad activity against Gram-negative bacteria, including multidrug-resistant (MDR) organisms like carbapenem-resistant Enterobacterales (CRE), carbapenem-resistant *Pseudomonas aeruginosa*, and *Acinetobacter baumannii*.
- The antibacterial activity of this molecule is due to its ability to achieve high periplasmic concentrations by hijacking the bacterial iron transport machinery, which increases cell entry.
 - In addition, cefiderocol remains stable to hydrolysis by serine β-lactamases (ESBLs, KPCs, and OXA-type carbapenemases) and MBL.
- In this study, the activities of cefiderocol and comparator agents were analyzed against carbapenem-nonsusceptible Enterobacterales, including molecularly characterized isolates, collected as part of the SENTRY Antimicrobial Surveillance Program for USA during 2020–2023.

Materials and Methods

Bacterial organisms

- This study comprised a collection of 15,147 Enterobacterales collected from various clinical specimens from patients hospitalized in 35 medical centers in all 9 US Census Divisions during 2020–2023. Only consecutive isolates (1 per patient infection episode) responsible for documented infections according to local criteria were included.
- Bacterial identification was confirmed by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following the Clinical and Laboratory Standards Institute (CLSI) M07 (2018) guidelines.
- Frozen-form broth microdilution panels were manufactured by JMI Laboratories (North Liberty, IA, USA) and contained cation-adjusted Mueller-Hinton broth for comparator agents.
- Susceptibility testing for cefiderocol used broth microdilution panels containing iron-depleted media per CLSI guidelines.
- Quality assurance was performed by sterility checks, colony counts, and testing CLSI-recommended quality control reference strains.
- MIC interpretations were performed using CLSI breakpoints for comparators and FDA/CLSI breakpoints for cefiderocol (≤4/8/≥16 mg/L for susceptible, intermediate, and resistant).
- Enterobacterales displaying MIC values ≥2 mg/L for imipenem (excluded for *P. mirabilis, P. penneri*, and indole-positive Proteeae) or meropenem were subjected to genome sequencing and screening of β-lactamase genes.

Screening of β-lactamase genes

- Selected isolates had total genomic DNA extracted by the fully automated Thermo Scientific™ KingFisher™ Flex Magnetic Particle Processor (Cleveland, OH, USA), which was used as input material for library construction.
- DNA libraries were prepared using the Nextera™ or Illumina DNA Prep™ library construction protocol (Illumina, San Diego, CA, USA) following the manufacturer's instructions and were sequenced on MiSeq or NextSeq Sequencer platforms at JMI Laboratories.
- FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.15.3. An in-house software was applied to align the assembled sequences against a comprehensive in-house database containing known β-lactamase genes.

Results

- A total of 1.0% (157/15,147) of all Enterobacterales isolates were not susceptible to imipenem and/or meropenem and 75.8% (119/157) carried carbapenemase genes (Figure 1 and Table 1).
 - Class A bla_{KPC} (72.3%; 86/119) prevailed, followed by class B MBL genes (17.6%; 21/119) and class D bla_{OXA-48} variants (5.9%; 7/119).
- Cefiderocol (96.6–98.3% susceptible) had MIC_{50} of 0.5 mg/L and MIC_{90} of 4 mg/L against carbapenem-non-susceptible and carbapenemase-carrying Enterobacterales; other agents had lower susceptibilities (6.7–86.5%) (Table 1).
- Cefiderocol (100% susceptible) and β-lactam-β-lactamase inhibitor (BL/BLI) combinations (96.6–100% susceptible) were active against Enterobacterales carrying class A carbapenemases, whereas only cefiderocol ($MIC_{50/90}$, 2/4 mg/L; 95.2% susceptible) had activity against those carrying class B genes (Table 1).
- Cefiderocol and ceftazidime-avibactam were active (100% susceptible for both) against Enterobacterales carrying class D genes (Table 1).
- Cefiderocol ($MIC_{50/90}$, 0.25/4 mg/L; 92.1% susceptible), meropenem-vaborbactam ($MIC_{50/90}$, 0.25/4 mg/L; 92.1% susceptible), and ceftazidime-avibactam ($MIC_{50/90}$, 0.5/8 mg/L; 94.7% susceptible) were the most active agents against carbapenem-nonsusceptible but carbapenemase-negative Enterobacterales (Table 1).

Figure 1. Distribution of carbapenemase genes[a] detected among carbapenem-nonsusceptible Enterobacterales

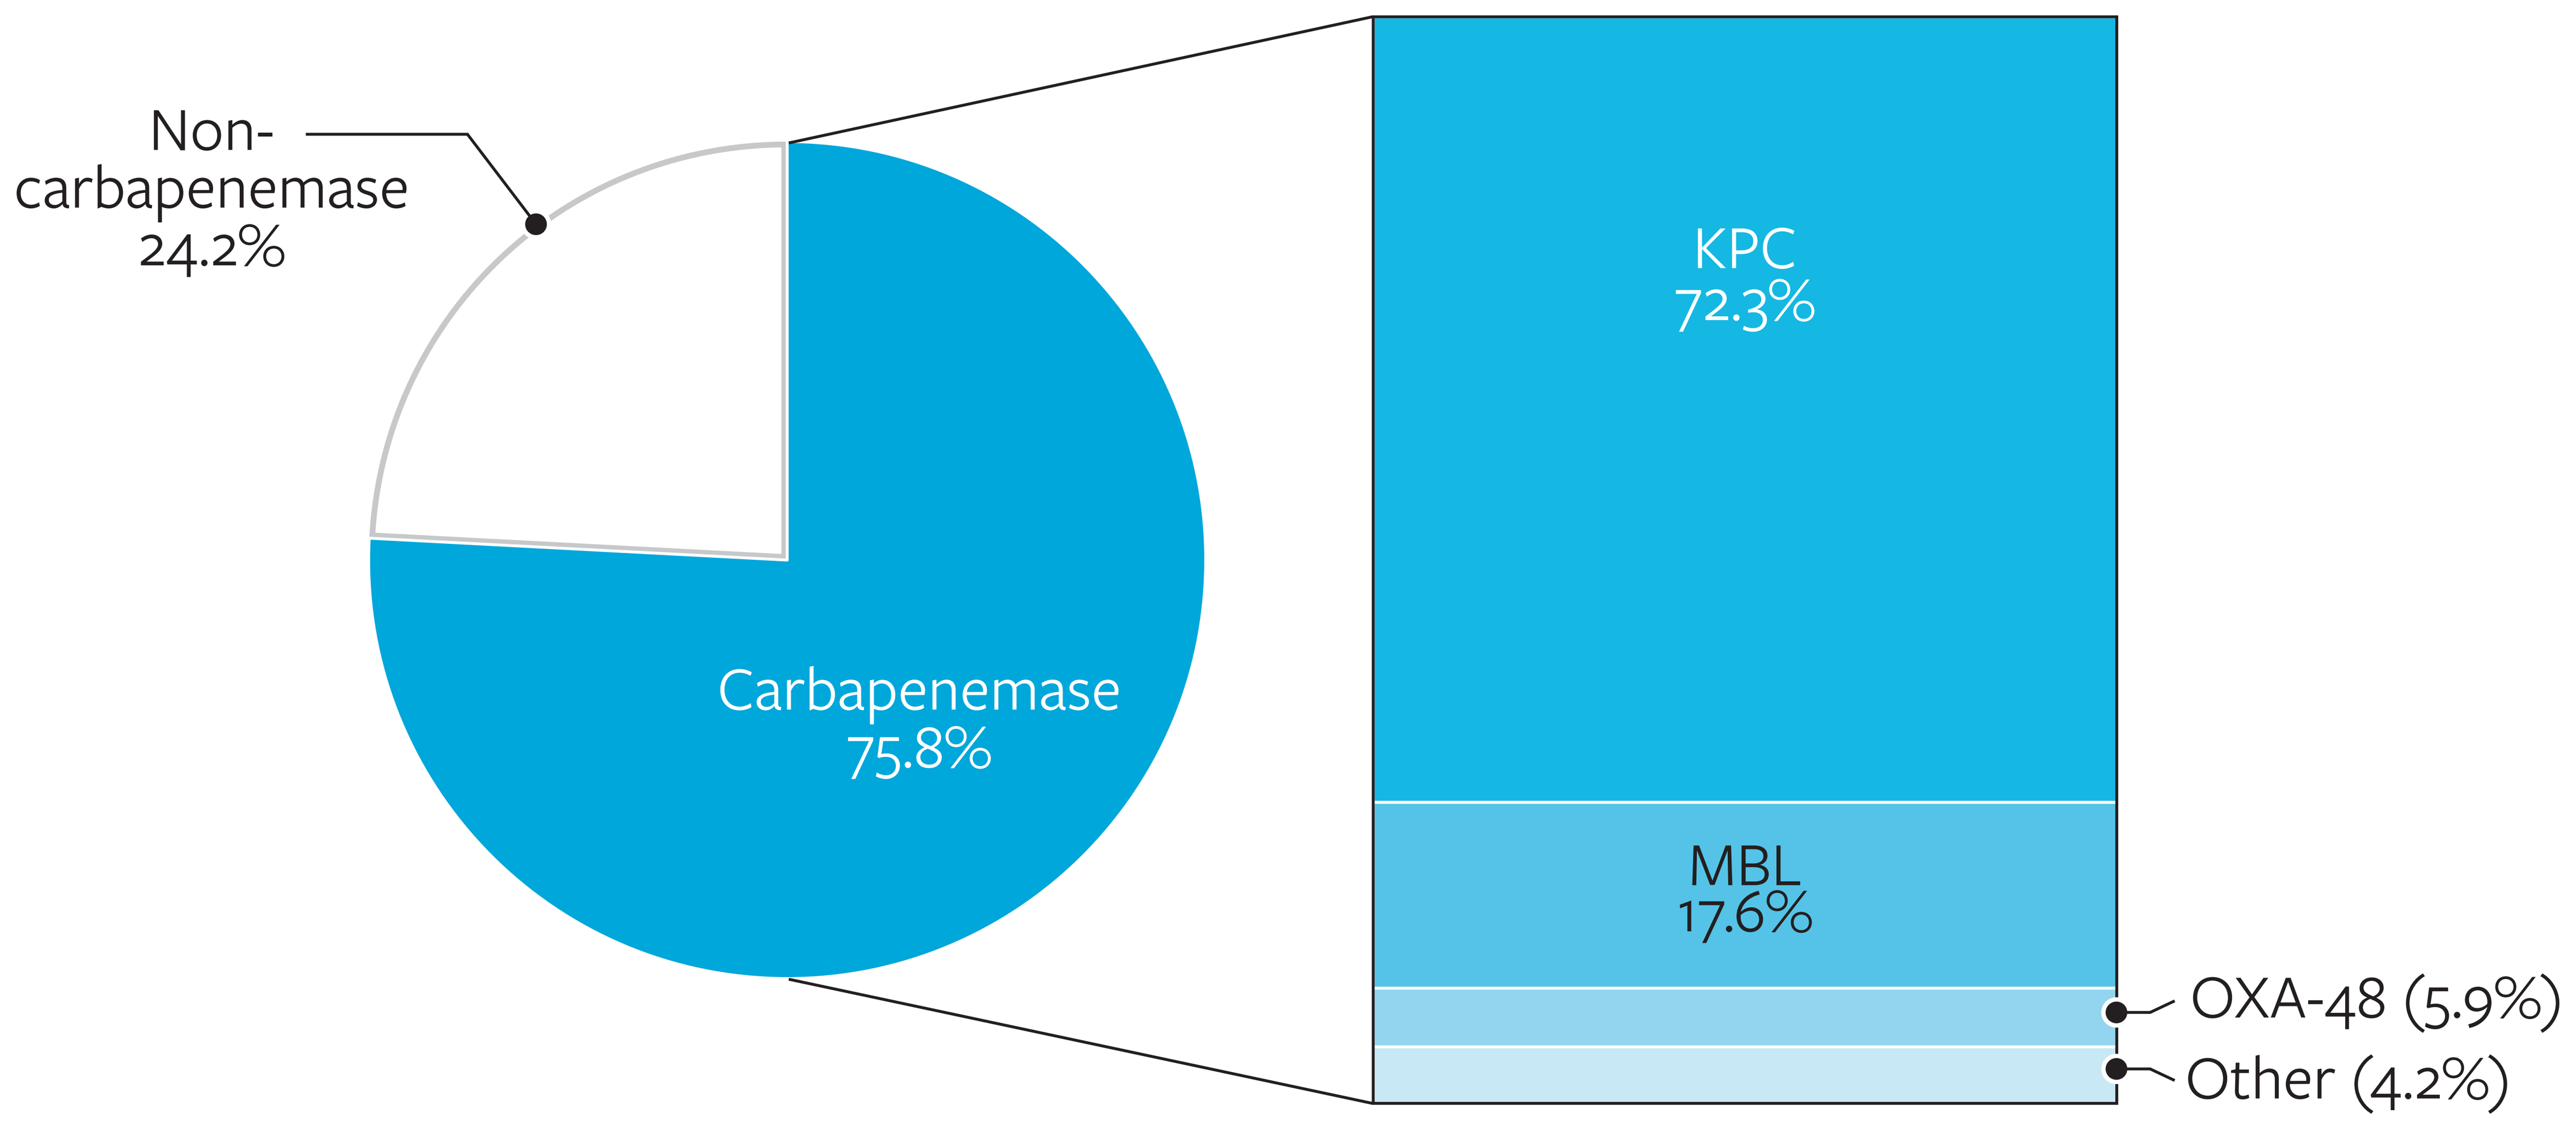
Non-
carbapenemase
24.2%
Carbapenemase
75.8%
KPC
72.3%
MBL
17.6%
OXA-48 (5.9%)
Other (4.2%)

[a] See footnotes of Table 1 for additional details related to carbapenemase alleles.

Table 1. Activity of cefiderocol and β-lactam-β-lactamase inhibitor combinations against Enterobacterales and resistant subsets from the USA

Phenotype/genotype[a] (No.)	MIC_{50}/MIC_{90} in mg/L (% susceptible by CLSI criteria)[b]					
	FDC	IMR	MEV	CZA	MER	COL
Carbapenem-nonS (157)	0.5/4 (96.6)	0.25/>8 (75.8)	0.12/>8 (83.1)	1/>32 (86.5)	4/>32 (24.2)	0.25/>8 (78.6)
Carbapenemase-positive[c] (119)	0.5/4 (98.3)	0.12/>8 (73.1)	0.06/>8 (76.5)	1/>32 (80.7)	16/>32 (6.7)	0.25/>8 (83.2)
Class A (89)	0.5/2 (100)	0.12/0.5 (96.6)	0.03/0.5 (98.9)	1/2 (100)	8/>32 (5.6)	0.25/>8 (83.1)
Class B (21)	2/4 (95.2)	>8/>8 (0.0)	>8/>8 (9.5)	>32/>32 (0.0)	32/>32 (9.5)	0.25/>8 (76.2)
Class D (7)	0.5/— (100)	4/— (14.3)	>8/— (14.3)	1/— (100)	>32/— (14.3)	0.12/— (100)
Carbapenemase-negative[d] (38)	0.25/4 (92.1)	0.5/2 (76.3)	0.25/4 (92.1)	0.5/8 (94.7)	0.5/8 (50.0)	0.25/>8 (57.9)

Abbreviations: FDC, cefiderocol; IMR, imipenem-relebactam; MEV, meropenem-vaborbactam; CZA, ceftazidime-avibactam; MER, meropenem; COL, colistin.
[a] Carbapenem-nonS, isolates non-susceptible to imipenem (excluded for *P. mirabilis, P. penneri*, and indole-positive Proteeae) and/or meropenem based on CLSI criteria (MIC values ≥2 mg/L).
[b] Cefiderocol MIC results were interpreted according to the CLSI criteria (equivalent to FDA). MIC for comparator agents was interpreted based on the CLSI criteria, except for colistin where the EUCAST susceptible breakpoint was applied.
[c] Includes the class A bla_{KPC-2} (36), bla_{KPC-3} (48), bla_{KPC-4} (2), bla_{SME-2} (2), and bla_{SME-4} (1) genes; the class B bla_{IMP-4} (1), bla_{NDM-1} (11), bla_{NDM-5} (6), bla_{NDM-7} (2), and bla_{VIM-1} (1) genes; and the class D bla_{OXA-48} (4), $bla_{OXA-181}$ (1), and $bla_{OXA-232}$ (2). It also includes the combinations bla_{NDM-5} + bla_{KPC-65} (1) and bla_{NDM-1} + $bla_{OXA-181}$ (1).
[d] Carbapenemase genes were not detected in the following species: *Citrobacter freundii* species complex (1), *Enterobacter cloacae* species complex (1), *Escherichia coli* (6), *Klebsiella oxytoca* (1), *K. pneumoniae* (10), *Raoultella ornithinolytica* (2), and *Serratia marcescens* (17).

Conclusions

- Cefiderocol activity against Enterobacterales was consistent, regardless of isolate phenotype or genotype.
 - Cefiderocol activity was observed against isolates carrying serine and MBL carbapenemases, and also against OXA-48-like, where approved β-lactam/β-lactamase inhibitor combinations showed limited activity.
- These data emphasize cefiderocol as an important option for the treatment of infections caused by resistant Enterobacterales, including carbapenem-nonsusceptible isolates.

Acknowledgements

This research and poster presentation were sponsored by Shionogi & Co., LTD.

References

1. Clinical and Laboratory Standards Institute. 2018. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. *M07 11th Edition*. Wayne, PA, USA.

2. Clinical and Laboratory Standards Institute. 2024. Performance standards for antimicrobial susceptibility testing. *M100 34th Edition*. Wayne, PA, USA.

3. FDA Susceptibility Test Interpretive Criteria: https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed April 2024.

4. Mendes RE, Jones RN, Woosley LN, Cattoir V, Castanheira M. 2019. Application of next-generation sequencing for characterization of surveillance and clinical trial isolates: Analysis of the distribution of β-lactamase resistance genes and lineage background in the United States. *Open Forum Infect Dis* 6: S69–S78.

5. Ong'uti S, Czech M, Robilotti E, Holubar M. 2021. Cefiderocol: A new cephalosporin stratagem against multidrug resistant Gram-negative bacteria. *Clin Infect Dis*. 74: 1303–1312.

6. Syed YY. 2021. Cefiderocol: A review in serious Gram-negative bacterial infections. *Drugs*. 24: 1–13.

Contact

Rodrigo E. Mendes, Ph.D., FIDSA
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo.mendes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2024_23-SHI-09_A4_Enteric_IDWeek2024.pdf

Charges may apply. No personal information is stored.

IDWeek 2024, October 16–19, 2024, Los Angeles, California